43065

Regional Variation in Alzheimer's Disease Progression in a Clinical Trial Setting

David Henley[1], Sherie Dowsett[1], Yun-Fei Chen[1], Hong Liu-Seifert[1], Joshua Grill[2], Rachelle Doody[3], Paul Aisen[4], Rema Raman[4], David Miller [5], Ann Marie Hake[1], Jeffrey Cummings[6]

[1]Lilly Research Laboratories, Indianapolis, IN, USA; [2]Mary Easton Center for Alzheimer's Disease Research, UCLA David Geffen School of Medicine, Los Angeles, CA, USA; [3]Baylor College of Medicine, Department of Neurology, Houston, TX, USA; [4]UC San Diego School of Medicine, San Diego, CA, USA; [5]Dementia and Geriatric Psychiatry, Bracket , Wayne PA, USA; [6]Cleveland Clinic Lou Ruvo Center for Brain Health, Las Vegas, NV, USA

BACKGROUND

- To facilitate enrollment and meet local registration requirements, sponsors have increasingly implemented multi-national Alzheimer's disease (AD) studies.
- Regional variability is expected but recognizing the extent of this can be helpful in planning AD study implementation.

AIM

- To aid researchers designing and implementing multi-national AD trials, we assessed disease progression across geographic regions using placebo data from 4 large, multi-national clinical trials of investigational compounds developed to target AD pathophysiology.

METHODS

- Similarly-designed, randomized, double-blind, placebo-controlled trials with nearly identical entry criteria enrolled patients aged ≥55 years with mild or moderate AD, based on the National Institute of Neurological and Communicative Disorders and Stroke /AD and Related Disorders Association (NINCDS/ADRDA) criteria for probable AD
 - Two semagacestat (IDENTITY) studies, each with an initial treatment period of 76 weeks (Doody 2013)
 - Two 80-week solanezumab (EXPEDITION) studies (Doody 2014)
- Disease progression was assessed as change from baseline to endpoint using cognitive, functional and global scales (see Table 3), administered in the local language(s).

Statistical Methods

- Descriptive analyses were performed for observed mean score and observed mean change in outcome from baseline at each scheduled visit.
- A Mixed Model Repeated Measures (MMRM) was used to estimate the regional means at each scheduled visit by adjusting for baseline score; region; education (<8 years, 8-12 years, >12 years); age at baseline; gender; APOE e4 status; MMSE stratification factor at Visit 1 (mild or moderate); and concomitant AChEI and/or Memantine use at baseline (yes or no).

RESULTS

ENROLLMENT AND STUDY COMPLETION (TABLE 1)

- Overall, data from 2079 subjects who were assigned to the placebo arm of one of the four studies (EXPEDITION and EXPEDITION2, n= 1025; IDENTITY and IDENTITY2, n=1054) were included in the analyses.
- NA accounted for most sites and subjects enrolled; fewest subjects were enrolled in AU.
- Some regions with relatively low enrollment included countries that participated only in the IDENTITY program (Belgium, Denmark, Finland, Serbia, India).
- The percentage of subjects who completed a study ranged from 56% (EE) to 84% (JP).

Table 1: Number of Sites, Enrolled Subjects and Subjects Completing the Study by Country and Region

Region / Country	No. Sites	No. Enrolled	% Completers[a]	
			EXPEDITION	IDENTITY
North America (NA)	**179**	**832**	**72**	**37**
US	154	714	71	41
Canada	25	118	80	19
Western Europe/Israel (WE)	**112**	**412**	**77**	**36**
Belgium	4	8	-	63
Denmark	2	5	-	60
Finland	3	6	-	100
France	16	69	92	20
Germany	23	104	76	29
Israel	7	22	-	36
Italy	18	74	74	6
Spain	16	48	63	50
Sweden	12	37	85	80
UK	11	39	72	36
South America /Mexico (SA)	**50**	**196**	**75**	**9**
Argentina	25	88	67	0
Brazil	14	67	85	0
Chile	7	17	-	47
Mexico	4	24	-	0
Eastern Europe/Russia (EE)	**49**	**195**	**61**	**4**
Bulgaria	5	18	-	6
Hungary	4	16	-	6
Poland	13	48	67	25
Romania	4	17	-	0
Russia	11	45	54	0
Serbia	3	8	-	0
Turkey	4	23	-	0
Ukraine	5	20	-	0
Japan (JP)	**46**	**191**	**88**	**21**
Asia (AS)	**40**	**169**	**84**	**0**
China	6	22	-	0
India	7	10	-	0
Korea	16	85	79	0
Taiwan	11	52	93	0
Australia/S. Africa (AU)	**25**	**84**	**84**	**39**
Australia	18	58	84	50
South Africa	7	26	-	31

[a]For EXPEDITION, completer defined as subject who had completed the 80-wk double-blind study period. For IDENTITY, completer defined as subject who had completed the 76-wk initial treatment period; denominator in this case was the no. of subjects who had opportunity to complete 76 wks of treatment *before* study drug was stopped at request of sponsor, and study was amended

Baseline Characteristics by Region (Table 2)

- Numerical differences in baseline characteristics were evident across regions
- AS had the lowest proportion of subjects with mild disease (36%)
- Generally, subjects were oldest in NA and SA, and youngest in WE and EE and had received most years of education in NA and fewest years in SA and AS; there were fewer males than females enrolled in all regions but proportionally more male subjects in WE and fewer in SA and EE; while 74-94% of subjects received concomitant AD treatment, it was most common in WE and least common in EE and AU; APOE ε4 carriers were most common in WE and NA and least common in AS.

Table 2: Baseline Characteristics by Region

	NA (n=832)	WE (n=412)	SA (n=196)	EE (n=195)	JA (n=191)	AS (n=169)	AU (n=84)	All Regions (N=2079)
Mild AD[a]	547 (66)	265 (64)	114 (58)	106 (54)	132 (69))	71 (36)	57 (68)	1292 (62)
Age[b]	75.0 (8.1)	71.6 (7.7)	74.6 (7.97)	70.9 (7.7)	73.1 (7.7)	72.2 (7.7)	73.0 (7.1)	73.4 (8.0)
Education[b]	14.00 (3.11)	11. 20 (4.17)	9.05 (4.51)	11.75 (3.72)	11. 76 (2.84)	9.64 (4.69	12.18 (3.38)	12.13 (4.06)
Male[a]	381 (45.8)	205 (49.8)	66 (33.7)	70 (35.9)	73 (38.2)	68 (40.2)	38 (45.2)	901 (43.3)
AChEI and/or memantine[a]	736 (88.5)	387 (93.9)	167 (85.2)	145 (74.4)	173 (90.6)	139 (82.2)	66 (78.6)	1813 (87.2)
APOE ε4 Carriers[a]	481 (63.0)	218 (65.7)	96 (51.1)	90 (51.4)	98 (53.0)	39 (42.4)	51 (61.4)	1073 (59.0)

[a]n (%)
[b]Mean (SD)

Disease Progression by Region (Table 3, Figure 1)

- At baseline, disease severity as measured by ADAS Cog, ADCS-ADL, and CDR-SB was worse for the EE population compared with populations of other regions; this was not the case for MMSE and NPI.
- Of all regional populations, EE showed the greatest cognitive and functional decline from baseline; JP, AS and/or SA showed the least cognitive and functional decline
- For ADAS Cog_{11} specifically, the EE population showed the most cognitive decline over the course of the study (mean change from baseline to 18m was 11.0) while AS and JP populations showed the least decline (mean change from baseline to 18m was 3.5 and 4.4, respectively); NA, AU, and WE populations showed similar decline (mean change from baseline to 18m, 6.0 to 7.5).
- For the SA population, the mean NPI score decreased substantially from Week 52 to Week 76/80

References:

(1) Doody RS, et al. 2013 NEJM 369(4):341-350. (2) Doody RS et al. 2014 NEJM 370(4):311-321.

Figure 1: Observed Mean Score at Each Time Point by Region for Outcome Measures

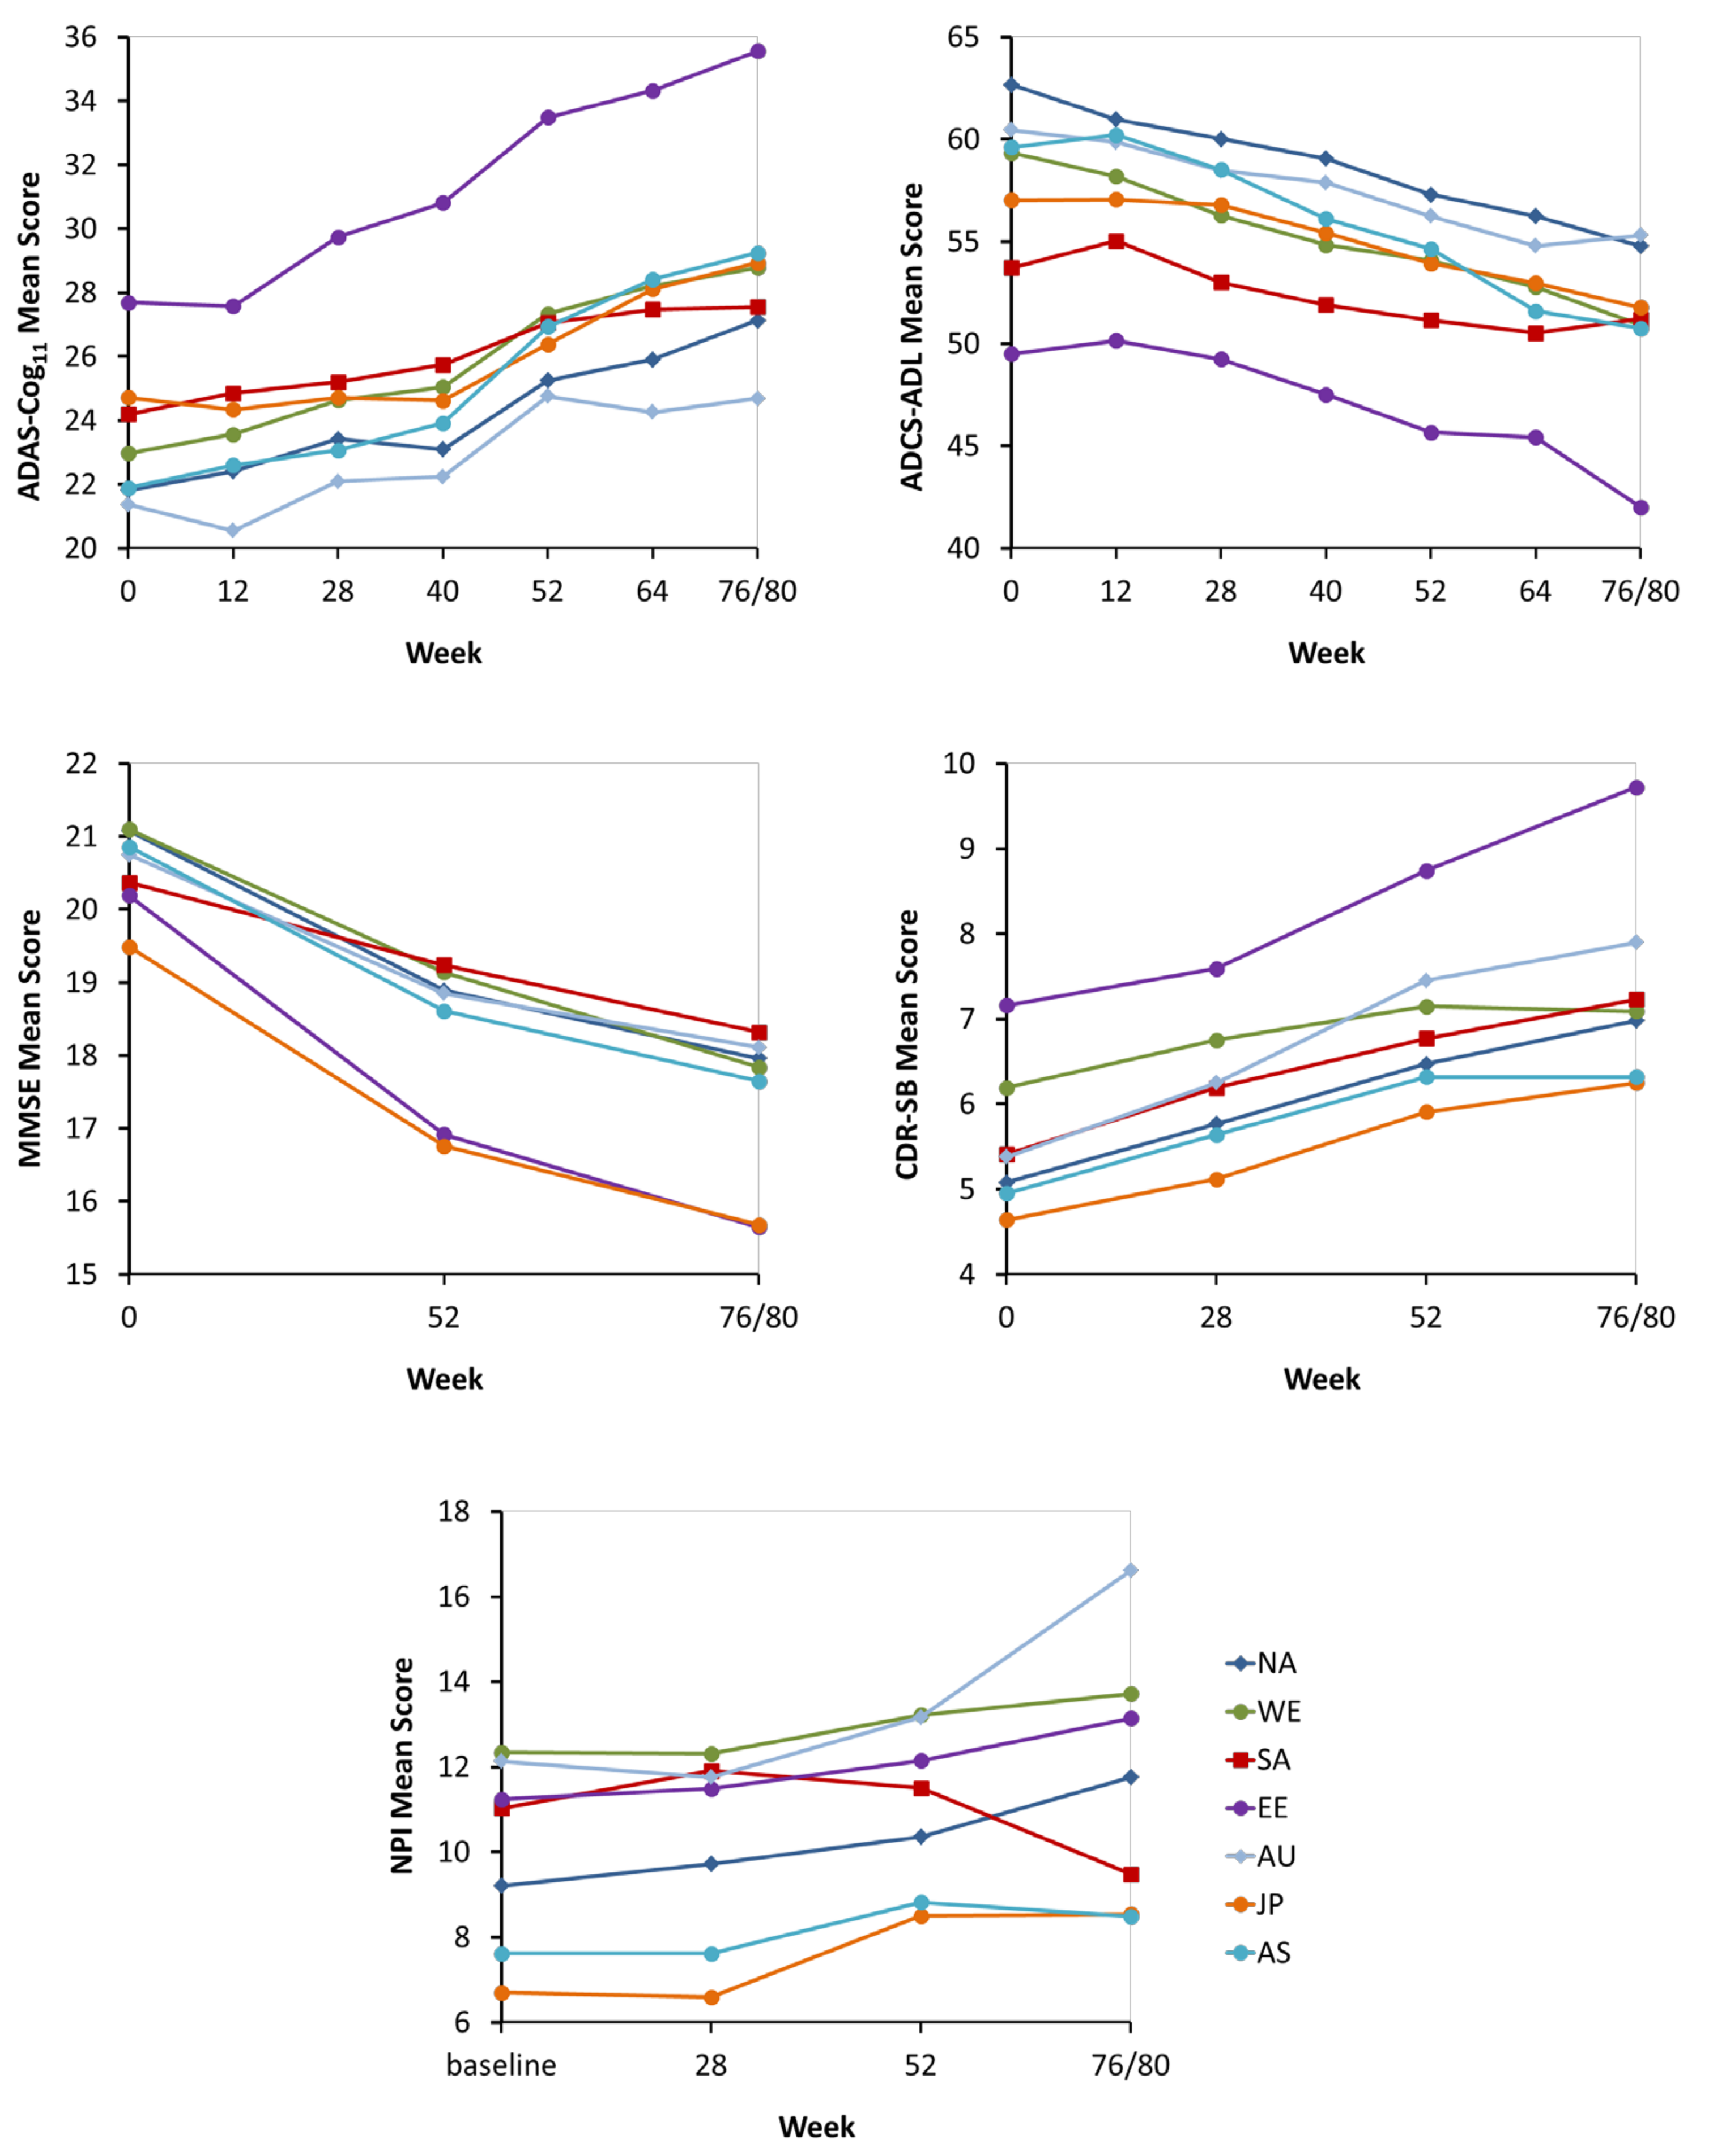
ADAS-Cog11 Mean Score
ADCS-ADL Mean Score
MMSE Mean Score
CDR-SB Mean Score
NPI Mean Score
Week
NA
WE
SA
EE
AU
JP
AS

ACKNOWLEDGEMENTS

We are grateful to the participants and investigators of the four trials, and to Laura Ramsey for poster preparation

Table 3: Observed Mean Change (SD) from Baseline to 76/80 Weeks By Region for Outcome Measures

	NA (n=832)	WE (n=412)	SA (n=196)	EE (n=195)	JP (n=191)	AS (n=169)	AU (n=84)	All Regions
ADAS Cog11 Baseline	21.81 (9.01)	22.97 (9.18)	24.20 (8.75)	27.69 (11.13)	21.37 (6.80)	24.72 (7.72)	21.89 (9.51)	23.02 (9.16)
76/80 wks	6.04 (9.44)	7.46 (9.68)	4.76 (8.41)	10.95 (10.77)	4.41 (7.99)	3.52 (7.98)	7.3 (11.54)	6.23 (9.48)
ADCS-ADL Baseline	62.68 (11.66)	59.34 (13.62)	53.72 (14.17)	49.51 (16.91)	60.45 (11.16)	57.02 (4.80)	59.62 (13.04)	59.16 (13.76)
76/80 wks	-9.16 (12.13)	-10.84 (13.44)	-5.57 (12.78)	-11.51 (14.16)	-5.94 (9.39)	-7.85 (9.75)	-9.00 (14.89)	-8.95 (12.48)
MMSE Baseline	21.08 (3.67)	21.10 (3.56)	20.37 (3.07)	20.19 (3.16)	20.75 (3.10)	19.49 (3.57)	20.86 (3.49)	20.77 (3.51)
76/80 wks	-3.39 (4.59)	-3.66 (4.70)	-2.52 (4.18)	-5.28 (5.95)	-2.78 (4.13)	-2.93 (4.06)	-3.45 (4.73)	-3.38 (4.60)
CDR-SB Baseline	5.08 (2.48)	5.41 (2.70)	6.19 (2.74)	7.16 (3.34)	4.95 (2.66)	4.64 (2.53)	5.38 (2.32)	5.41 2.74)
76/80 wks	6.98 (3.91)	7.23 (3.98)	7.09 (3.59)	9.72 (4.66)	6.32 (3.92)	6.25 (3.25)	7.90 (4.32)	7.10 (3.96)
NPI Baseline	9.21 (10.94)	11.03 (11.73)	12.34 (12.82)	11.24 (11.86)	6.70 (8.69)	7.62 (8.68)	12.13 (10.80)	9.81 (11.13)
76/80 wks	2.86 (13.36)	2.97 (13.98)	-1.80 (14.08)	2.30 (12.57)	2.41 (8.95)	1.83 (10.00)	5.29 (14.11)	2.47 (13.11)

MMRM Analysis Findings (Baseline to Endpoint LS Mean Change)

- ADASCog11: EE, AU and WE exhibited the greatest change (9.0, 9.0 and 8.9, JP and AS the smallest (3.5 and 3.7).
- ADCS-ADL: NA and WE exhibited the greatest change (11.4 and 13.4), JP, AS and SA the smallest (6.5, 7.0 and 7.9).
- MMSE: Changes ranged from 2.06 (JP) to 4.32 (EE)
- CDR-SB: EE exhibited the greatest change (3.42), SA, AS, and JP the smallest (1.25 to 1.48)
- NPI: Findings were variable across regions , similar to the observed mean change data (Fig 1)

DISCUSSION/CONCLUSIONS

- Limitations: Some regional populations were small in number; geographic groupings were based on expected similarities in genetic and/or environmental (e.g., healthcare, culture) factors across countries within region, but groupings may still be somewhat arbitrary
- There was heterogeneity across regions in baseline and endpoint clinical measures.
- These findings suggest differences among regions in measure of disease progression (e.g., due to differences in assessment or disease progression) or baseline differences in disease severity
- These initial data may be helpful to researchers planning multinational AD trials.
- Further exploration is warranted in assessing potential regional differences in disease progression.

Alzheimer's Association International Conference; Copenhagen, Denmark; July 12 – 17, 2014

Copyright © 2014 Eli Lilly and Company

Sponsored by Eli Lilly and Company