SNR06629B Poster no abstract:Layout 1 27/5/10 14:55 Page 1

P456

20th Meeting of the European Neurological Society (ENS), Berlin, Germany, 19–23 June 2010

ASSOCIATIONS BETWEEN ANNUALIZED RELAPSE RATE AND LONG-TERM EXPOSURE TO SUBCUTANEOUS INTERFERON BETA-1A IN THE PREVENTION OF RELAPSES AND DISABILITY BY INTERFERON BETA-1A SUBCUTANEOUSLY IN MULTIPLE SCLEROSIS (PRISMS) LONG-TERM FOLLOW-UP STUDY

B Uitdehaag,[1] C Constantinescu,[2] P Cornelisse,[3] D Jeffery,[4] L Kappos,[5] D Li,[6] M Sandberg-Wollheim,[7] A Traboulsee,[6] E Verdun,[3] V Rivera[8] for the PRISMS study group

[1]MS Center, Department of Neurology, VU University Medical Center, Amsterdam, the Netherlands; [2]Division of Clinical Neurology, University of Nottingham, UK; [3]Merck Serono S.A. – Geneva, Switzerland; [4]Department of Neurology, Wake Forest University School of Medicine, Winston-Salem, USA; [5]Department of Neurology, University Hospital, Basel, Switzerland; [6]Departments of Radiology and Medicine (Neurology), University of British Columbia, Vancouver, Canada; [7]Department of Neurology, University Hospital, Lund, Sweden; [8]Department of Neurology, Baylor College of Medicine, Houston, Texas, USA*

Introduction

- The placebo-controlled phase of the PRISMS (Prevention of Relapses and disability by Interferon beta-1a Subcutaneously in Multiple Sclerosis) study demonstrated the efficacy of interferon (IFN) beta-1a, 44 or 22 mcg subcutaneously (sc) three times weekly (tiw), on clinical and radiological outcomes in patients with relapsing–remitting multiple sclerosis (RRMS).[1]
- Data over 4 years showed that clinical and radiological benefits were maintained for both doses, with evidence of dose–response favouring the high-dose regimen, and greater benefits seen in patients who were randomized to active treatment from the start ('early-start' patients).[2]
- Data from a long-term follow-up (LTFU) visit at around 8 years after study start demonstrated the continued clinical and magnetic resonance imaging (MRI) benefit of sc IFN beta-1a, with more pronounced treatment effects seen in early-start patients.[3]
- The LTFU dataset provides an opportunity to examine the hypothesis that cumulative exposure to disease-modifying drug (DMD) therapy is an important predictor of treatment outcomes.
- Previous *post hoc* analyses of these data have shown that patients with the highest cumulative time receiving sc IFN beta-1a and continuous treatment have better clinical[4,5] and MRI[5] outcomes for up to 8 years, compared with patients with lower cumulative exposure or interruptions in treatment.
- We conducted exploratory *post hoc* analyses to investigate annualized relapse rates (ARRs) in patients with RRMS receiving sc IFN beta-1a for up to 8 years according to total cumulative time on therapy, total cumulative dose exposure and continuous versus non-continuous treatment.

Methods

Study design

- The PRISMS study comprised the phases outlined below.[3–5]
 - In the initial 2-year, double-blind phase, patients with RRMS were randomized to receive IFN beta-1a, 44 or 22 mcg sc tiw, or placebo (years 1–2).
 - After 2 years, patients originally randomized to placebo were then re-randomized to IFN beta-1a, 44 or 22 mcg sc tiw, for an additional 2 years (years 3–4).
 - All patients were given the choice of continuing to receive blinded or open-label treatment for a further 2 years (years 5–6).
 - After year 6, patients could continue treatment with any or no DMD.
- Patients were eligible for enrolment in the LTFU study if they had been randomized to treatment in the original PRISMS study, regardless of whether they had subsequently withdrawn from the study.
- Patients had a single LTFU assessment close to the seventh or eighth anniversary of their baseline visit.
 - The assessment included a neurological evaluation as well as a retrospective review of data (including treatment exposure, neurological status and disease progression) collected since the year-4 assessment.[3]

Post hoc exploratory analyses

- LTFU data from patients randomized to the three original study arms were pooled and ranked into quartiles from lowest to highest for a) cumulative time on sc IFN beta-1a and b) cumulative dose exposure. Calculations of both parameters took into account periods when patients were not receiving sc IFN beta-1a.
- ARRs across 2-year intervals were assessed in the minimum (lowest quartile, MIN) and maximum (highest quartile, MAX) cumulative time and dose groups.
- ARRs were also calculated in patients initially randomized to IFN beta-1a, 44 mcg sc tiw, and receiving 'continuous' or 'non-continuous' (interrupted) therapy.
 - The 'continuous' subgroup consisted of all patients who had been randomized on study day 1 to IFN beta-1a, 44 mcg sc tiw, and who had remained on that dose until the LTFU visit, with no interruptions and no other DMDs taken.
 - The 'non-continuous' subgroup consisted of all patients who had been randomized to IFN beta-1a, 44 mcg sc tiw, and who had some medication interruptions, irrespective of other DMDs taken.
- Only descriptive statistics were applied because no inferential statistical comparisons were made.

Results

- Of 560 patients originally randomized in the PRISMS study, 493 were available for participation in the LTFU study (three sites did not enter the LTFU study). Of these patients, 382 participated in the LTFU visit.
 - Of these 382 patients, 136 (35.6%) had originally been randomized to IFN beta-1a, 44 mcg sc tiw, 123 (32.2%) to IFN beta-1a, 22 mcg sc tiw, and 123 (32.2%) to placebo.

Cumulative time groups

- Mean (standard deviation; SD) cumulative time exposure was:
 - 156.8 (87.7) weeks in the MIN cumulative time group (n=96)
 - 390.3 (5.9) weeks in the MAX cumulative time group (n=95).
- Mean (SD) cumulative dose exposure was:
 - 13.7 (9.4) mg/patient in the MIN cumulative time group
 - 38.3 (11.0) mg/patient in the MAX cumulative time group.
- The MIN cumulative time group comprised patients originally randomized to IFN beta-1a, 44 or 22 mcg sc tiw, or placebo, whereas the MAX cumulative time group comprised only patients originally randomized to IFN beta-1a, 44 or 22 mcg sc tiw (**Table 1**).
- Patients in the MAX cumulative time group had a lower mean (SD) ARR than those in the MIN cumulative time group, from baseline to LTFU (0.51 [0.49] and 0.76 [0.55], respectively).
- Patients in the MAX cumulative time group also had a lower mean ARR than those in the MIN cumulative time group over each study period analysed (**Figure 1**).

Table 1. *MIN and MAX cumulative time and dose groups by original PRISMS study randomization groups.*

		Cumulative time on sc IFN beta-1a		Cumulative dose of sc IFN beta-1a	
	Cohort	**MIN**	**MAX**	**MIN**	**MAX**
Original PRISMS study randomization group, n (% of exposure cohort)	IFN beta-1a, 44 mcg sc tiw	21 (21.9)	53 (55.8)	14 (14.6)	91 (95.8)
	IFN beta-1a, 22 mcg sc tiw	22 (22.9)	42 (44.2)	28 (29.2)	0
	Placebo	53 (55.2)	0	54 (56.3)	4 (4.2)
	Total	96 (100)	95 (100)	96 (100)	95 (100)

IFN, interferon; MAX, highest quartile; MIN, lowest quartile; PRISMS, Prevention of Relapses and disability by Interferon beta-1a Subcutaneously in Multiple Sclerosis; sc, subcutaneous; tiw, three times weekly.

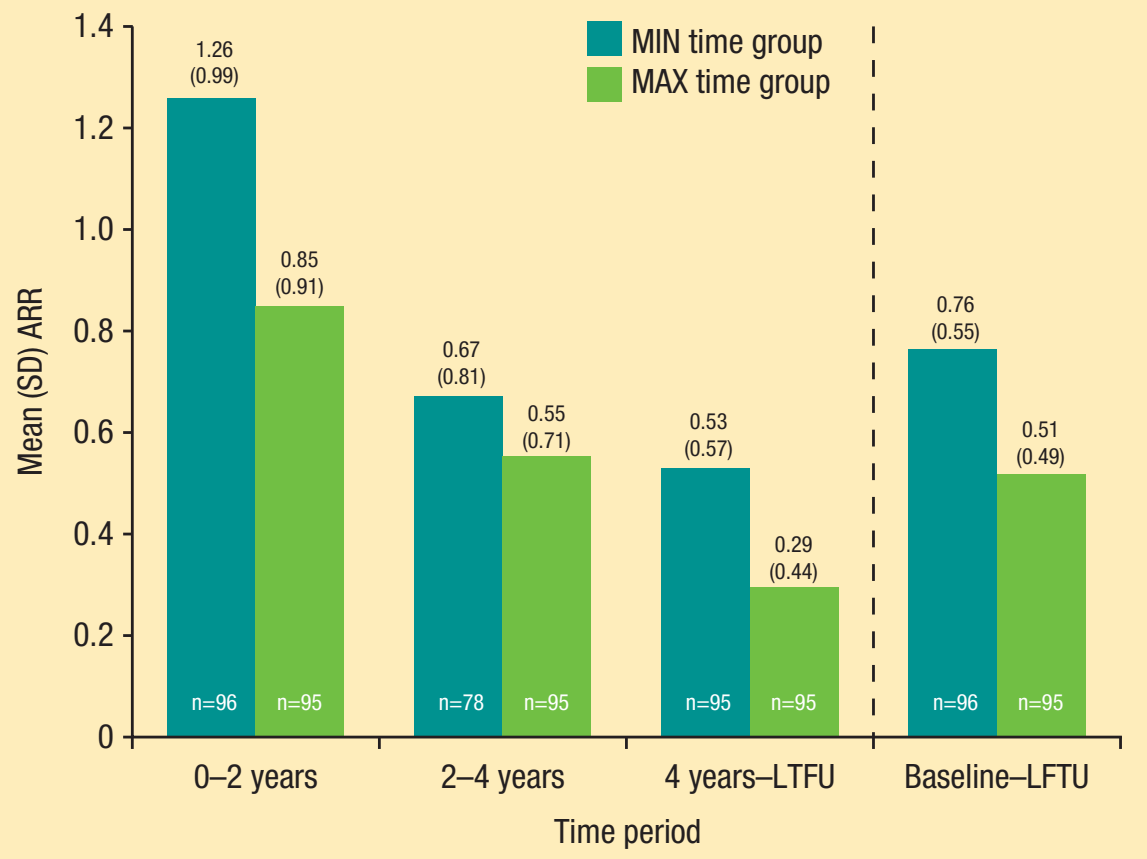
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0
Mean (SD) ARR
MIN time group
MAX time group
1.26 (0.99)
0.85 (0.91)
0.67 (0.81)
0.55 (0.71)
0.53 (0.57)
0.29 (0.44)
0.76 (0.55)
0.51 (0.49)
n=96 n=95
n=78 n=95
n=95 n=95
n=96 n=95
0–2 years
2–4 years
4 years–LTFU
Baseline–LFTU
Time period

ARR, annualized relapse rate; LTFU, long-term follow up; MAX, highest time quartile; MIN, lowest time quartile; SD, standard deviation.

Figure 1. *ARR in MIN and MAX groups for cumulative length of time on subcutaneous interferon beta-1a.*

Cumulative dose groups

- Mean (SD) cumulative dose exposure was:
 - 10.8 (6.0) mg/patient in the MIN cumulative dose group (n=96)
 - 46.6 (4.6) mg/patient in the MAX cumulative dose group (n=95).
- Mean (SD) cumulative time exposure was:
 - 172.4 (102.6) weeks in the MIN cumulative dose group
 - 375.3 (26.4) weeks in the MAX cumulative dose group.
- The MIN cumulative dose group mainly comprised patients originally randomized to placebo or the lower dose of IFN beta-1a (n=82/96, 85.4%), whereas the MAX cumulative dose group predominantly comprised patients originally randomized to the higher dose of IFN beta-1a (n=91/95, 95.8%; **Table 1**).
- Patients in the MAX cumulative dose group had a lower mean (SD) ARR than those in the MIN cumulative dose group, from baseline to LTFU (0.52 [0.51] and 0.72 [0.55], respectively).
- Patients in the MAX cumulative dose group also had a lower mean ARR than those in the MIN cumulative dose group over each study period analysed (**Figure 2**).

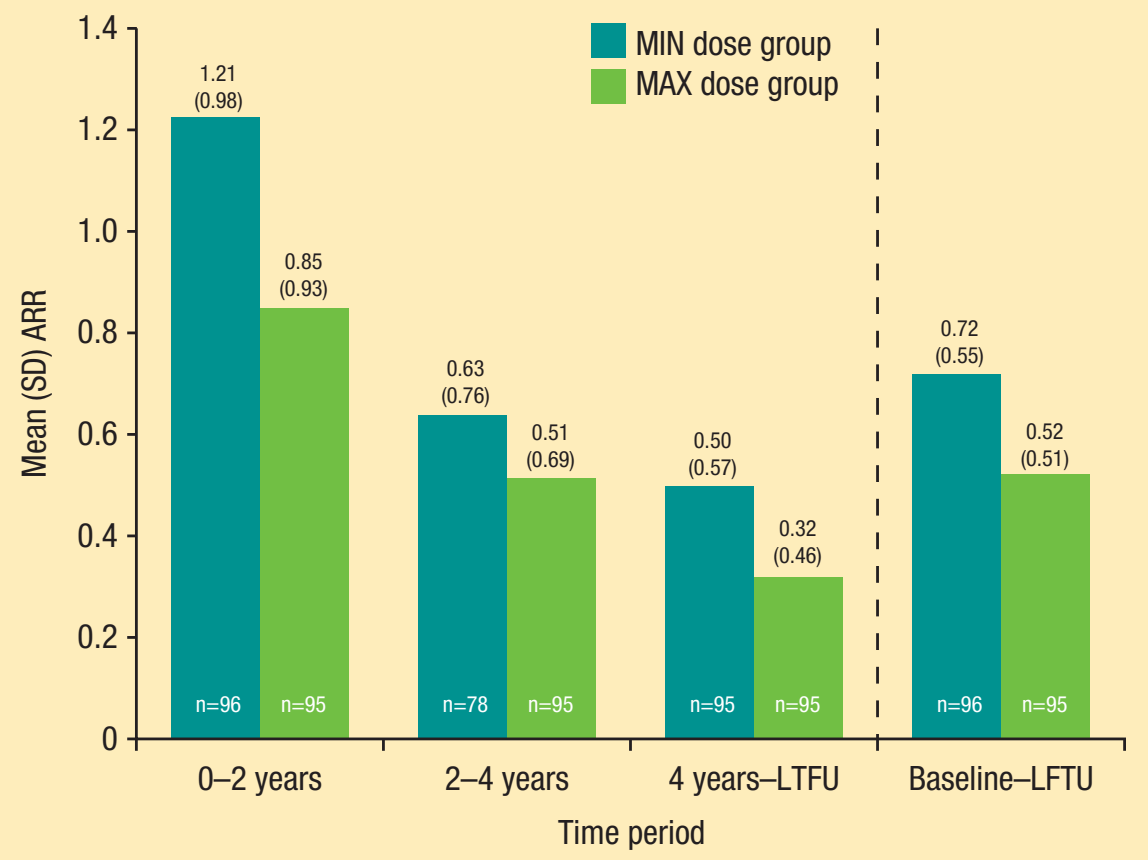
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0
Mean (SD) ARR
MIN dose group
MAX dose group
1.21 (0.98)
0.85 (0.93)
0.63 (0.76)
0.51 (0.69)
0.50 (0.57)
0.32 (0.46)
0.72 (0.55)
0.52 (0.51)
n=96 n=95
n=78 n=95
n=95 n=95
n=96 n=95
0–2 years
2–4 years
4 years–LTFU
Baseline–LFTU
Time period

ARR, annualized relapse rate; LTFU, long-term follow up; MAX, highest time quartile; MIN, lowest time quartile; SD, standard deviation.

Figure 2. *ARR in MIN and MAX groups for cumulative dose exposure to subcutaneous interferon beta-1a.*

Non-continuous and continuous groups

- Mean (SD) cumulative dose exposure was:
 - 34.0 (13.5) mg/patient in the non-continuous group (n=91)
 - 49.4 (2.6) mg/patient in the continuous group (n=45).
- Patients in the continuous group had a lower mean ARR than those in the non-continuous group, from baseline to LTFU (0.51 [0.54] and 0.61 [0.56], respectively).
- Patients in the continuous group also had a lower mean ARR than those in the non-continuous group over each study period analysed (**Figure 3**).

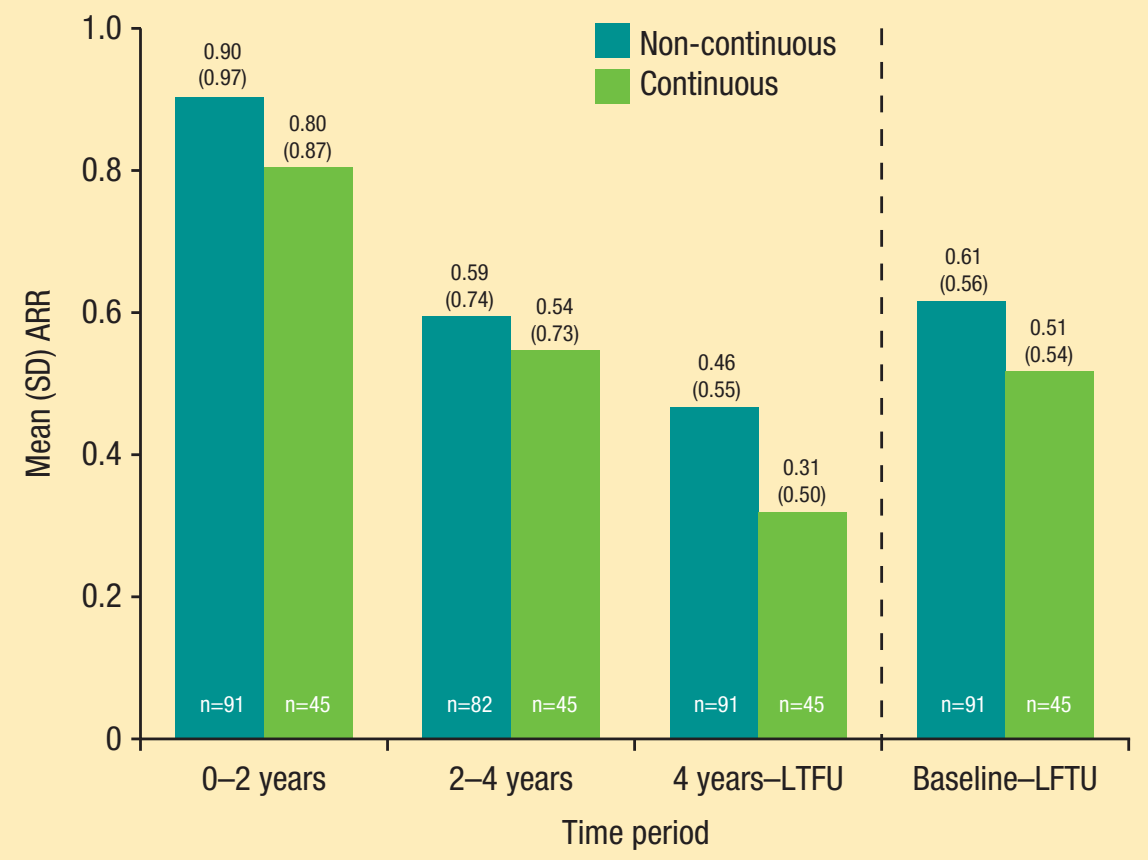
1.0
0.8
0.6
0.4
0.2
0
Mean (SD) ARR
Non-continuous
Continuous
0.90 (0.97)
0.80 (0.87)
0.59 (0.74)
0.54 (0.73)
0.46 (0.55)
0.31 (0.50)
0.61 (0.56)
0.51 (0.54)
n=91 n=45
n=82 n=45
n=91 n=45
n=91 n=45
0–2 years
2–4 years
4 years–LTFU
Baseline–LFTU
Time period

ARR, annualized relapse rate; LTFU, long-term follow up; SD, standard deviation.

Figure 3. *ARR in patients originally randomized to interferon beta-1a, 44 mcg subcutaneously three times weekly, with (non-continuous) or without (continuous) treatment interruptions (from baseline to LTFU).*

Conclusions

- Results from these *post hoc* exploratory analyses of data from the PRISMS LTFU study show that, in patients with RRMS, the greatest reductions on ARR were observed with the highest cumulative time and dose exposures to sc IFN beta-1a, over the long term (for up to 8 years).
- Lower ARRs were also seen in patients receiving continuous treatment with IFN beta-1a, 44 mcg sc tiw, for up to 8 years, than in those who had treatment interruptions.
- Although this analysis shows a correlation between treatment cessation and disease progression, the direction of causality cannot be established and other possible contributing factors may be involved. However, these findings support the importance of high levels of treatment exposure in patients with MS receiving IFN beta-1a and the maintenance of uninterrupted treatment over the long term.

References

1. PRISMS Study Group. Lancet 1998;352:1498–504.
2. PRISMS Study Group, University of British Columbia MS/MRI Analysis Group. Neurology 2001;56:1628–36.
3. Kappos L *et al*. Neurology 2006;67:944–53.
4. Uitdehaag B. J Neurol 2009;256(Suppl. 2):S122–3. Abstract P346.
5. Uitdehaag B. J Neurol 2009;256(Suppl. 2):S123. Abstract P347.

Acknowledgements

This study was supported by Merck Serono S.A. – Geneva, Switzerland.*
The authors thank the PRISMS study investigators and also Reza Sayeed for Caudex Medical (supported by Merck Serono S.A. – Geneva, Switzerland*) for assistance in the preparation of this poster.

Disclosures

B Uitdehaag has received honoraria for consultancy from Novartis, Merck Serono S.A. and Danone Research for consultancy activities. C Constantinescu has received compensation from Teva (advisory board and research support) and Merck Serono, Biogen Idec, Novartis and UCB (research support). P Cornelisse and E Verdun are employees of Merck Serono S.A. – Geneva, Switzerland.* D Jeffery has received research support and honoraria for speaking and consulting from Bayer, Biogen, Teva, Serono, Pfizer, GSK, Accorda and Novartis. L Kappos has received research support and honoraria or consultancy fees from Accorda Therapeutics Inc., Actelion Pharmaceuticals Ltd, Allozyne, BaroFold, Bayer Healthcare Pharmaceuticals, Bayer Schering Pharma, Bayhill, Biogen Idec, Boehringer Ingelheim, Eisai, Elan, Genmab, GlaxoSmithKline, Merck Serono, MedicNova, Novartis, Sanofi-Aventis, Santhera Pharmaceuticals, Shire PLC, Roche, Teva, UCB and others. D Li is the Director of the UBC MS/MRI Research Group which has been contracted to perform central analysis of MRI scans for therapeutic trials with Angiotech, Bayer, Berlex-Schering, Bio-MS, Centocor, Daiichi Sankyo, Genentech, Hoffmann-LaRoche, Merck-Serono, Perceptives, Schering-Plough, Teva Neurosciences, Sanofi-Aventis and Transition Therapeutics. He has also acted as a consultant to Genzyme and Novartis. M Sandberg-Wollheim has received honoraria from Serono Symposia International Foundation (lectures), Merck Serono (lectures, work in data safety monitoring boards [DSMBs]), Genentech (DSMB), Elan (advisory board), SEB Enskilda (lectures), honorarium for serving on the board of directors of Active Biotech in Lund, Sweden and honoraria as an external reviewer of a PhD thesis at University of Copenhagen. A Traboulsee has received personal compensation from Merck Serono for DSMB membership and for co-chairing a symposium, Bayer Healthcare for speaking, Teva Neurosciences for chairing a symposium, Neura as a member of an editorial advisory board and Daiichi for DSMB membership. V Rivera has received grants/honoraria from Bayer Healthcare, Biogen Idec, EMD Serono, Novartis, Stendhal Neuroscience, Teva, National Multiple Sclerosis Society and the Consortium of MS Centers.

*An affiliate of Merck KGaA, Darmstadt, Germany.