Dimebon Improves Cognition, Function, and Behavior in Mild and Moderate Alzheimer's Disease: Results by Severity of a One-Year, Double-Blind, Placebo-Controlled Study

Rachelle Doody,[1] Svetlana Gavrilova,[2] Ronald Thomas,[3] Paul Aisen,[4] Sergey Bachurin,[2] Lynn Seely,[5] and David Hung[5]

[1]Baylor College of Medicine, Houston, TX, United States, [2]Russian Academy of Medical Sciences, Moscow, Russian Federation, [3]University of California, San Diego, La Jolla, CA, United States, [4]Georgetown University Hospital, Washington, DC, United States, [5]Medivation, Inc, San Francisco, CA, United States

BACKGROUND

- Dimebon is an orally available investigational drug shown to have neuroprotective effects in models relevant to Alzheimer's disease (AD) and Huntington's disease.
- Dimebon improved cognition, functional ability, and behavior as compared to placebo in a 1-year, double-blind, placebo-controlled study of patients with mild-to-moderate AD.[1]
- The most potent mechanism of action for Dimebon identified to date is enhancement of mitochondrial function in the setting of cellular stress.

METHODS

- The study design and methods have been previously published.[1]
- 183 patients with mild-to-moderate AD (MMSE of 10-24) were randomized to receive Dimebon 20 mg TID or matching placebo for 26 weeks. After the initial 26 weeks, patients were offered the opportunity to remain on their blinded study drug for an additional 26 weeks (1-year double-blind total treatment period).
- Standard inclusion and exclusion criteria were utilized, although patients enrolled were not on any background anti-dementia therapy.
- Efficacy endpoints included the Alzheimer's Disease Assessment Scale—cognitive subscale (ADAS-cog); Clinician's Interview-Based Impression of Change, plus Caregiver Input (CIBIC-plus); Alzheimer's Disease Cooperative Study—Activities of Daily Living (ADCS-ADL); Mini-Mental State Examination (MMSE); and Neuropsychiatric Inventory (NPI).
- For the mild and moderate subgroup analyses, patients with mild AD were defined as those with a baseline MMSE > 18 (n = 94) and patients with moderate AD as those with a baseline MMSE ≤ 18 (n = 89).
- The data are presented using an observed-case analysis for the patient population that initially enrolled into the trial (n = 183) and also for the population of patients who consented for the entire 1-year treatment period (1-year cohort, n = 134).

Figure 2. ADAS-cog: Dimebon Significantly Improved Cognition at 1 Year in Patients With Mild and With Moderate AD

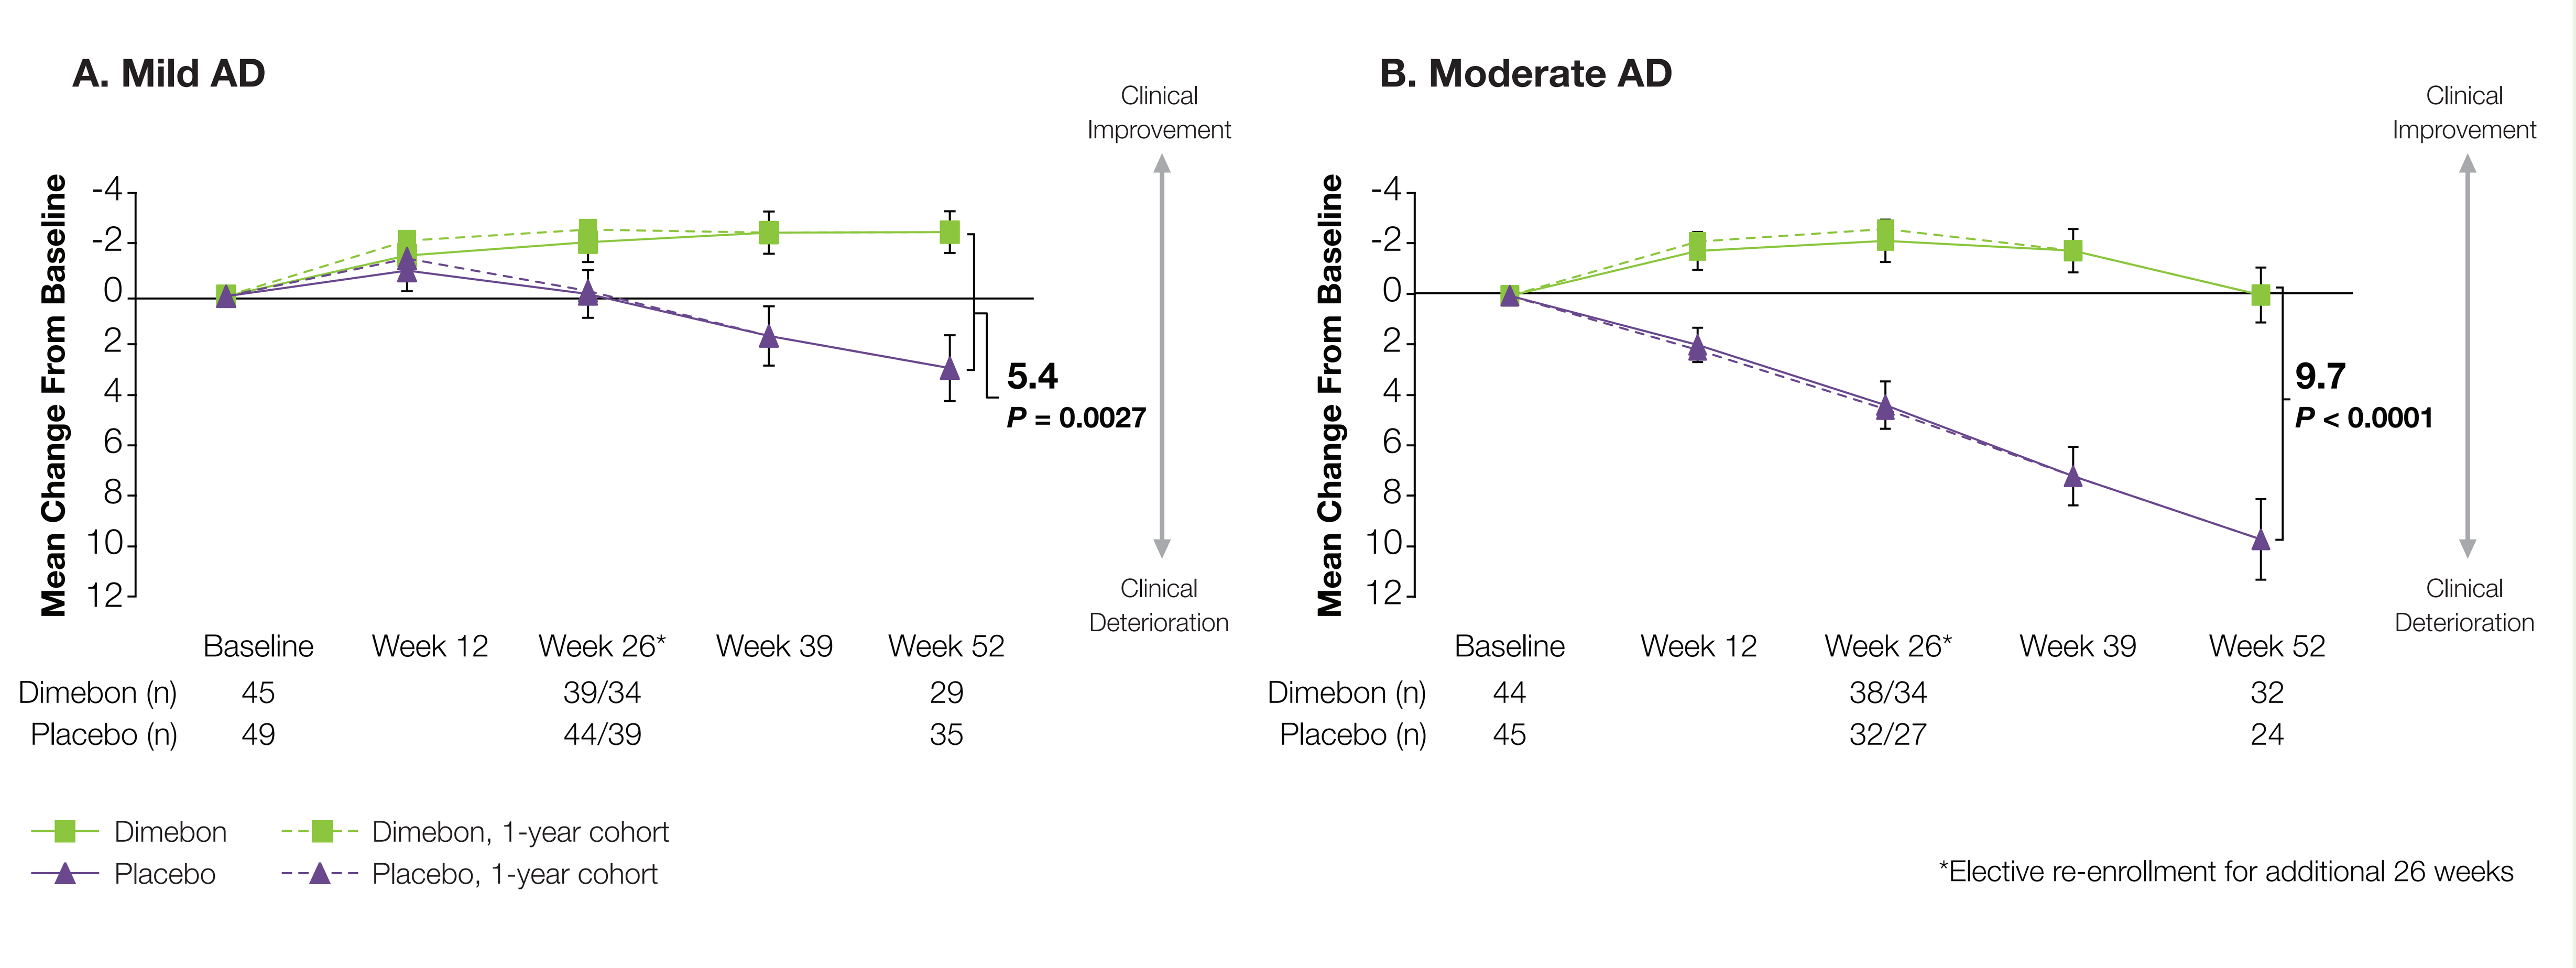
A. Mild AD
B. Moderate AD
Mean Change From Baseline
Clinical Improvement
Clinical Deterioration
5.4
P = 0.0027
9.7
P < 0.0001
Baseline
Week 12
Week 26*
Week 39
Week 52
Dimebon (n) 45 39/34 29
Placebo (n) 49 44/39 35
Dimebon (n) 44 38/34 32
Placebo (n) 45 32/27 24
Dimebon
Placebo
Dimebon, 1-year cohort
Placebo, 1-year cohort
*Elective re-enrollment for additional 26 weeks

Figure 4. ADLs: Dimebon Improved Activities of Daily Living in Patients With Mild and With Moderate AD

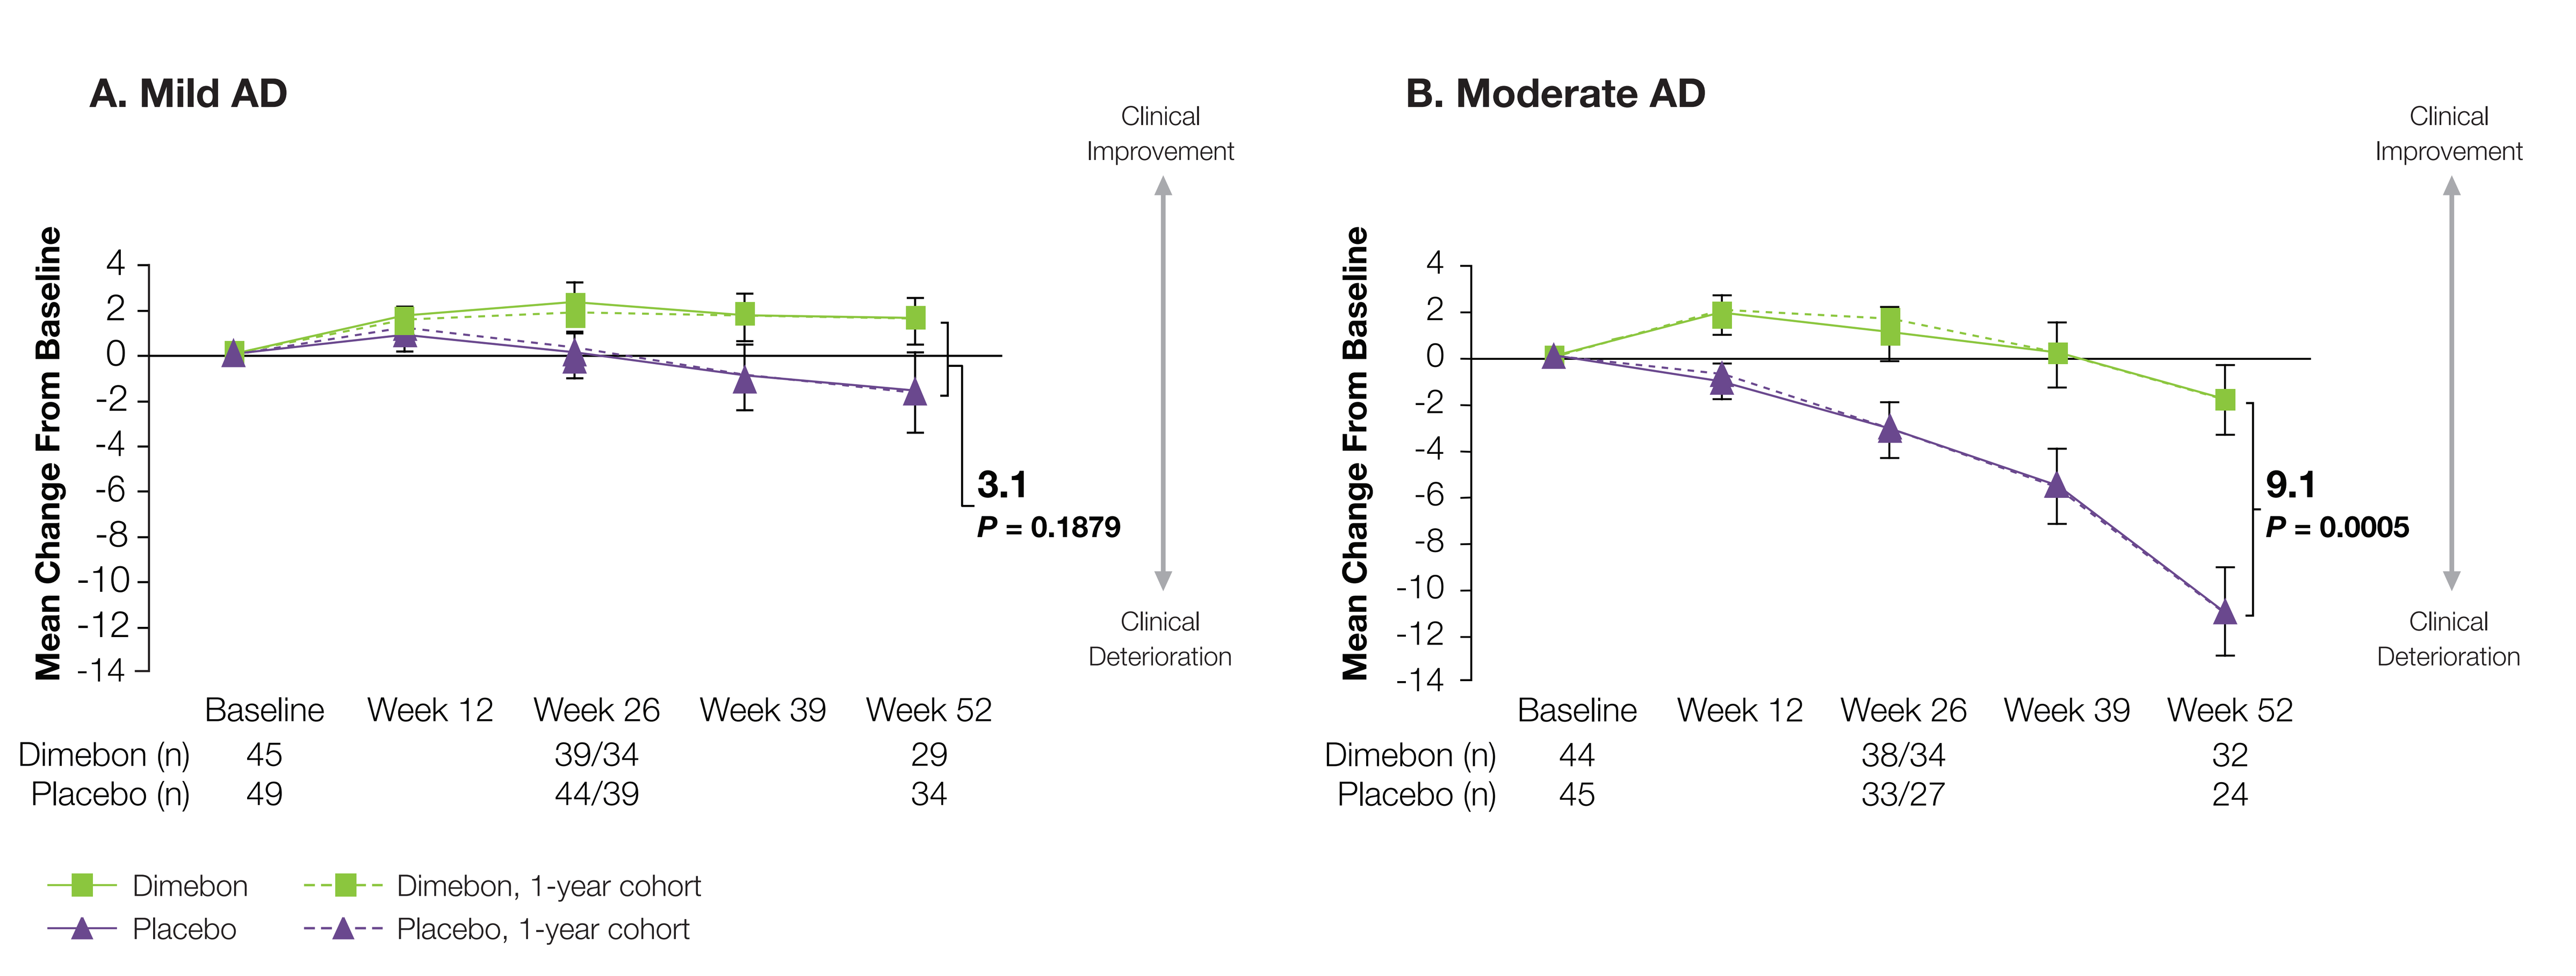
A. Mild AD
B. Moderate AD
Mean Change From Baseline
Clinical Improvement
Clinical Deterioration
3.1
P = 0.1879
9.1
P = 0.0005
Baseline
Week 12
Week 26
Week 39
Week 52
Dimebon (n) 45 39/34 29
Placebo (n) 49 44/39 34
Dimebon (n) 44 38/34 32
Placebo (n) 45 33/27 24
Dimebon
Placebo
Dimebon, 1-year cohort
Placebo, 1-year cohort

Figure 1. CIBIC-plus: Dimebon Improved Overall Function at 1 Year in Patients With Mild and With Moderate AD

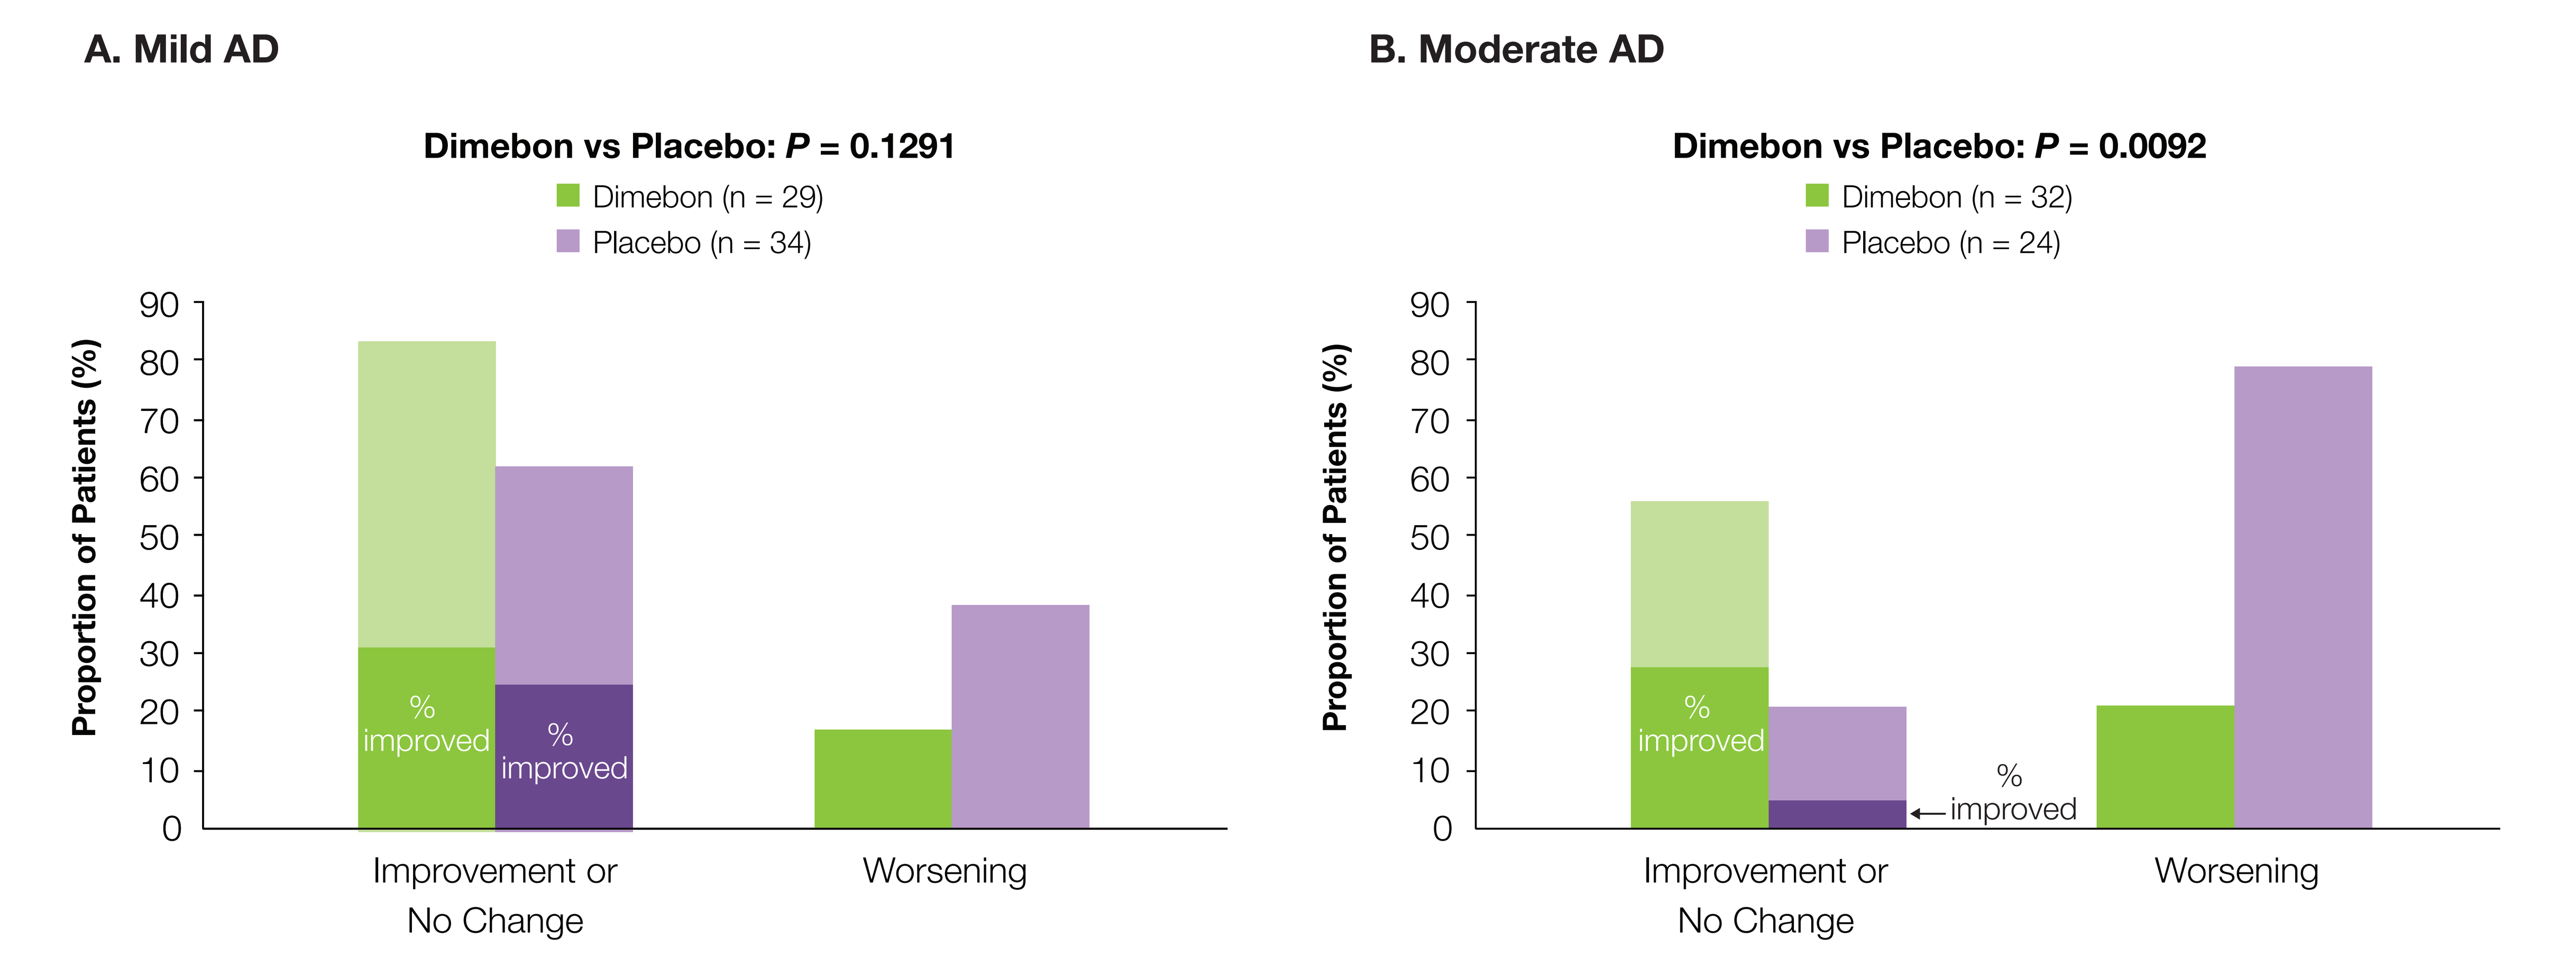
A. Mild AD
B. Moderate AD
Dimebon vs Placebo: P = 0.1291
Dimebon (n = 29)
Placebo (n = 34)
Dimebon vs Placebo: P = 0.0092
Dimebon (n = 32)
Placebo (n = 24)
Proportion of Patients (%)
% improved
Improvement or No Change
Worsening

Figure 3. NPI: Dimebon Significantly Improved Neuropsychiatric Symptoms in Patients With Moderate AD

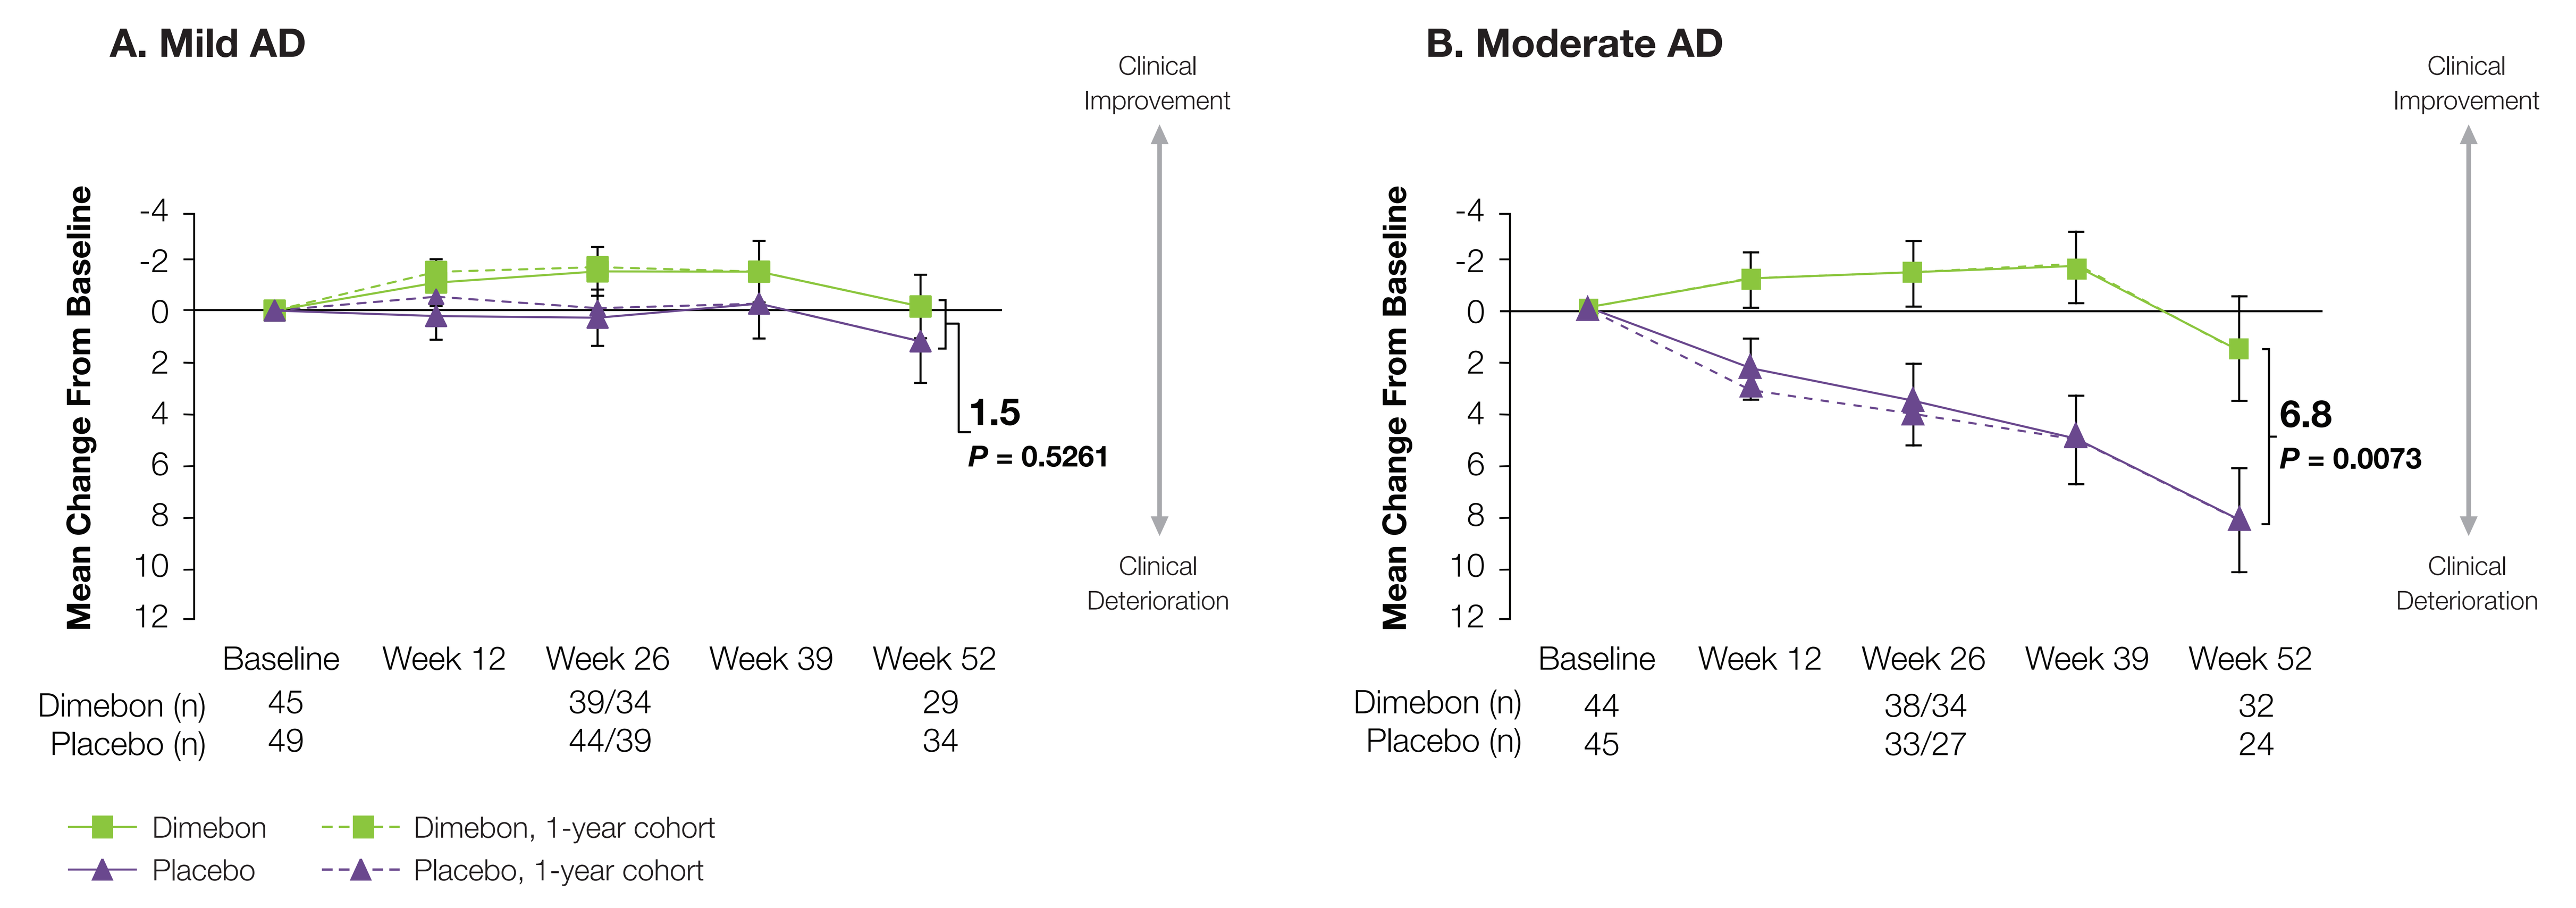
A. Mild AD
B. Moderate AD
Mean Change From Baseline
Clinical Improvement
Clinical Deterioration
1.5
P = 0.5261
6.8
P = 0.0073
Baseline
Week 12
Week 26
Week 39
Week 52
Dimebon (n) 45 39/34 29
Placebo (n) 49 44/39 34
Dimebon (n) 44 38/34 32
Placebo (n) 45 33/27 24
Dimebon
Placebo
Dimebon, 1-year cohort
Placebo, 1-year cohort

Table 1. Dimebon Treatment Was Well Tolerated

Week 52	Dimebon (n = 89)	Placebo (n = 94)
Number of patients reporting at least 1 adverse event, no. (%)	70 (79%)	70 (75%)
Number of subjects reporting at least 1 serious adverse event, no. (%)	3 (3%)	11 (12%)

CONCLUSIONS

- Patients with both mild and with moderate AD demonstrated benefit from treatment with Dimebon on cognition and memory, activities of daily living, behavior, and global function.
- Patients with mild AD were at or above baseline on all 5 outcome measures studied after 1 year of treatment.
 - Mild AD patients treated with Dimebon remained above baseline for 1 year on the ADAS-cog.
- Patients with moderate AD demonstrated substantial and sustained benefit over placebo on all 5 outcome measures at 1 year.
 - Moderate AD patients treated with Dimebon were improved 9.7 points over placebo on the ADAS-cog.
- Dimebon was safe and well tolerated.
- Dimebon improves the clinical course of patients with both mild and with moderate AD.

REFERENCE

1. Doody RS, Gavrilova SI, Sano M, et al; Dimebon investigators. Effect of dimebon on cognition, activities of daily living, behaviour, and global function in patients with mild-to-moderate Alzheimer's disease: a randomised, double-blind, placebo-controlled study. *Lancet*. 2008;372:207-215.

Dimebon Improves Cognition, Function, and Behavior in Mild and Moder
of a One-Year, Double-Blind, Placebo-Co

Rachelle Doody,[1] Svetlana Gavrilova,[2] Ronald Thomas,[3] Paul Aisen,[4] Sergey Bachurin,[2] Ly

[1]Baylor College of Medicine, Houston, TX, United States, [2]Russian Academy of Medical Sciences, Moscow, Russian Federation, [3]University of California, San Diego, La Jolla, CA, United States, [4]Georgetown

BACKGROUND

- Dimebon is an orally available investigational drug shown to have neuroprotective effects in models relevant to Alzheimer's disease (AD) and Huntington's disease.
- Dimebon improved cognition, functional ability, and behavior as compared to placebo in a 1-year, double-blind, placebo-controlled study of patients with mild-to-moderate AD.[1]
- The most potent mechanism of action for Dimebon identified to date is enhancement of mitochondrial function in the setting of cellular stress.

METHODS

- The study design and methods have been previously published.[1]
- 183 patients with mild-to-moderate AD (MMSE of 10-24) were randomized to receive Dimebon 20 mg TID or matching placebo for 26 weeks. After the initial 26 weeks, patients were offered the opportunity to remain on their blinded study drug for an additional 26 weeks (1-year double-blind total treatment period).
- Standard inclusion and exclusion criteria were utilized, although patients enrolled were not on any background anti-dementia therapy.
- Efficacy endpoints included the Alzheimer's Disease Assessment Scale—cognitive subscale (ADAS-cog); Clinician's Interview-Based Impression of Change, plus Caregiver Input (CIBIC-plus); Alzheimer's Disease Cooperative Study—Activities of Daily Living (ADCS-ADL); Mini-Mental State Examination (MMSE); and Neuropsychiatric Inventory (NPI).
- For the mild and moderate subgroup analyses, patients with mild AD were defined as those with a baseline MMSE > 18 (n = 94) and patients with moderate AD as those with a baseline MMSE ≤ 18 (n = 89).
- The data are presented using an observed-case analysis for the patient population that initially enrolled into the trial (n = 183) and also for the population of patients who consented for the entire 1-year treatment period (1-year cohort, n = 134).

Figure 2. ADAS-cog: Dimebon Significantly Improved Cognition at 1 Year in Patients With Mild and With Moderate AD

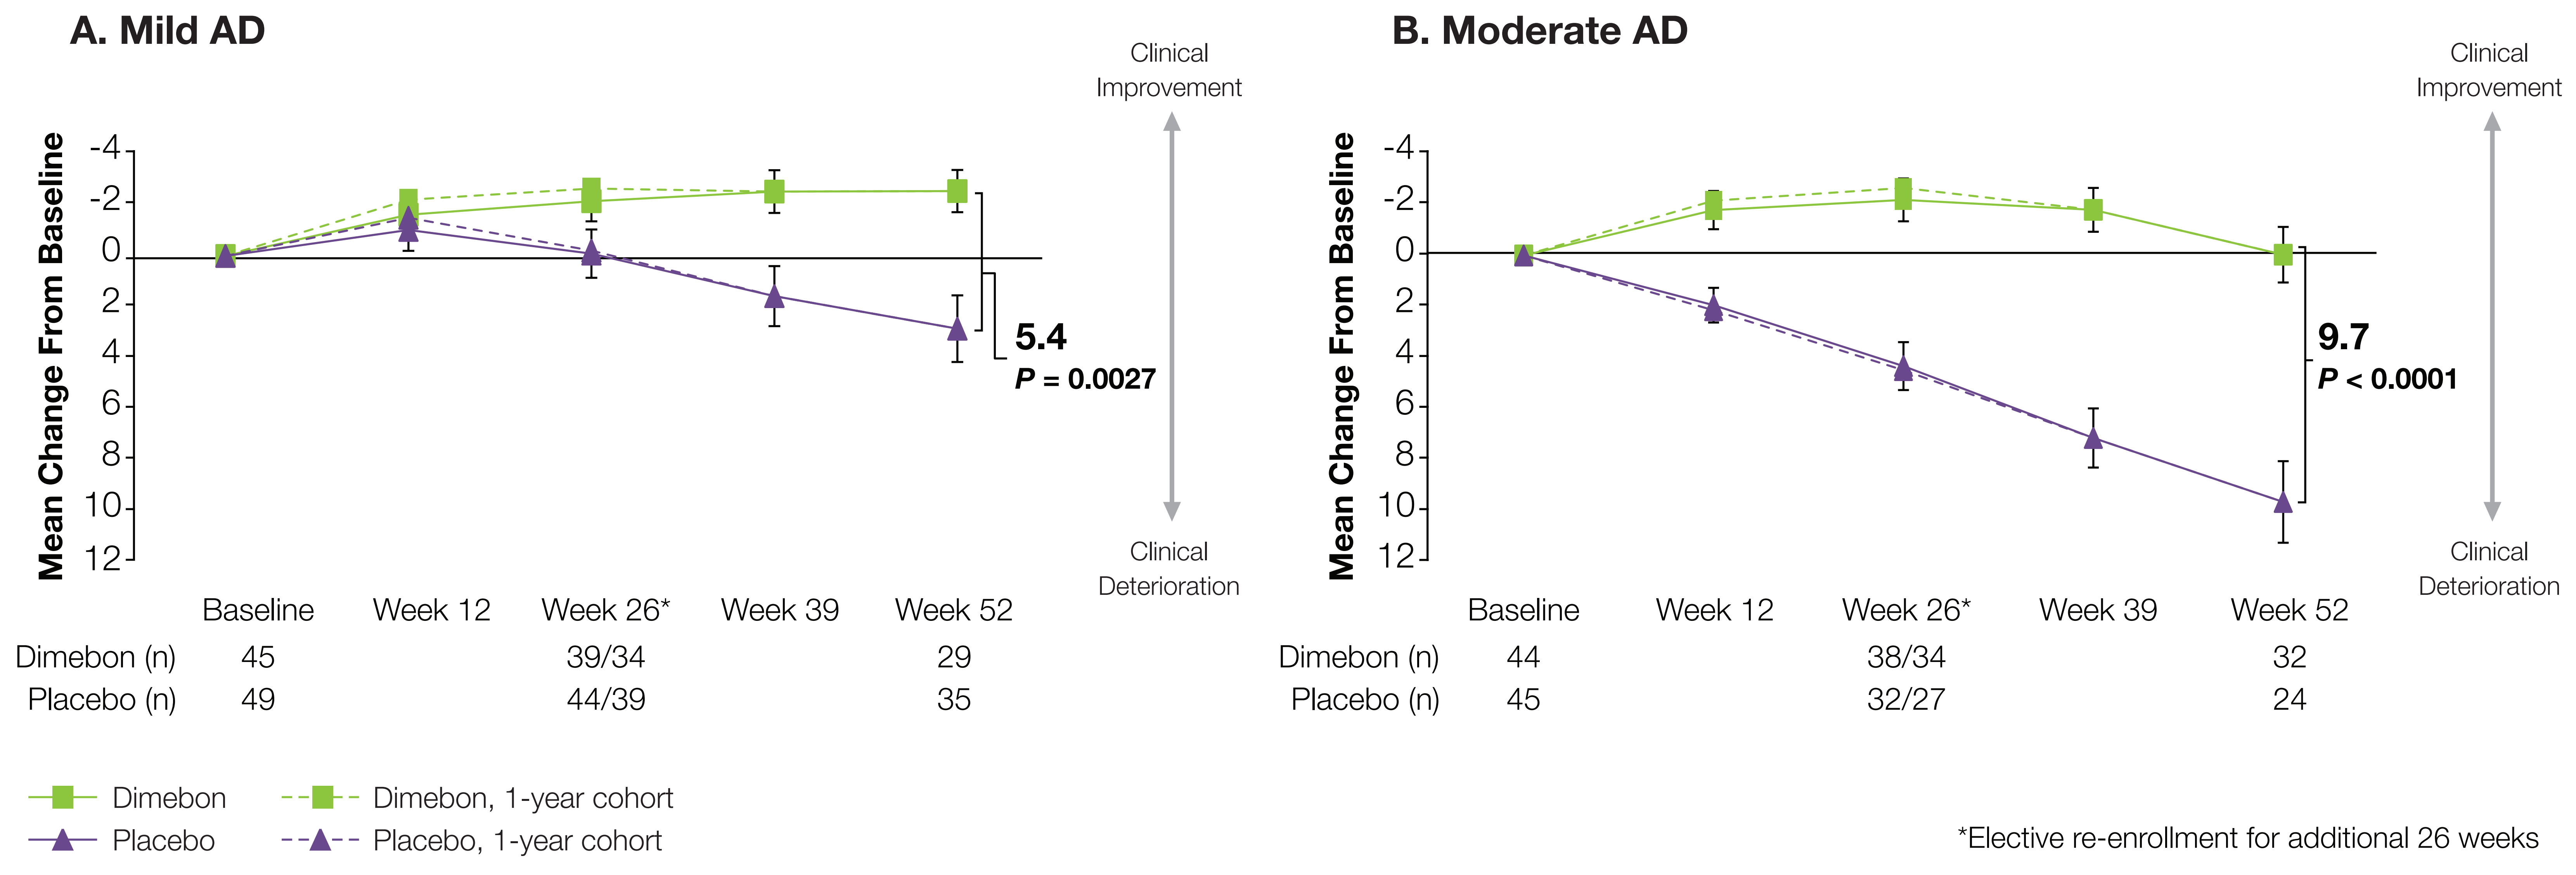
A. Mild AD
Clinical Improvement
Clinical Deterioration
Mean Change From Baseline
-4
-2
0
2
4
6
8
10
12
5.4
P = 0.0027
Baseline
Week 12
Week 26*
Week 39
Week 52
Dimebon (n) 45 39/34 29
Placebo (n) 49 44/39 35
B. Moderate AD
Clinical Improvement
Clinical Deterioration
Mean Change From Baseline
9.7
P < 0.0001
Dimebon (n) 44 38/34 32
Placebo (n) 45 32/27 24
Dimebon
Placebo
Dimebon, 1-year cohort
Placebo, 1-year cohort

*Elective re-enrollment for additional 26 weeks

erate Alzheimer's Disease: Results by Severity ontrolled Study

Lynn Seely,[5] and David Hung[5]

own University Hospital, Washington, DC, United States, [5]Medivation, Inc, San Francisco, CA, United States

Figure 4. ADLs: Dimebon Improved Activities of Daily Living in Patients With Mild and With Moderate AD

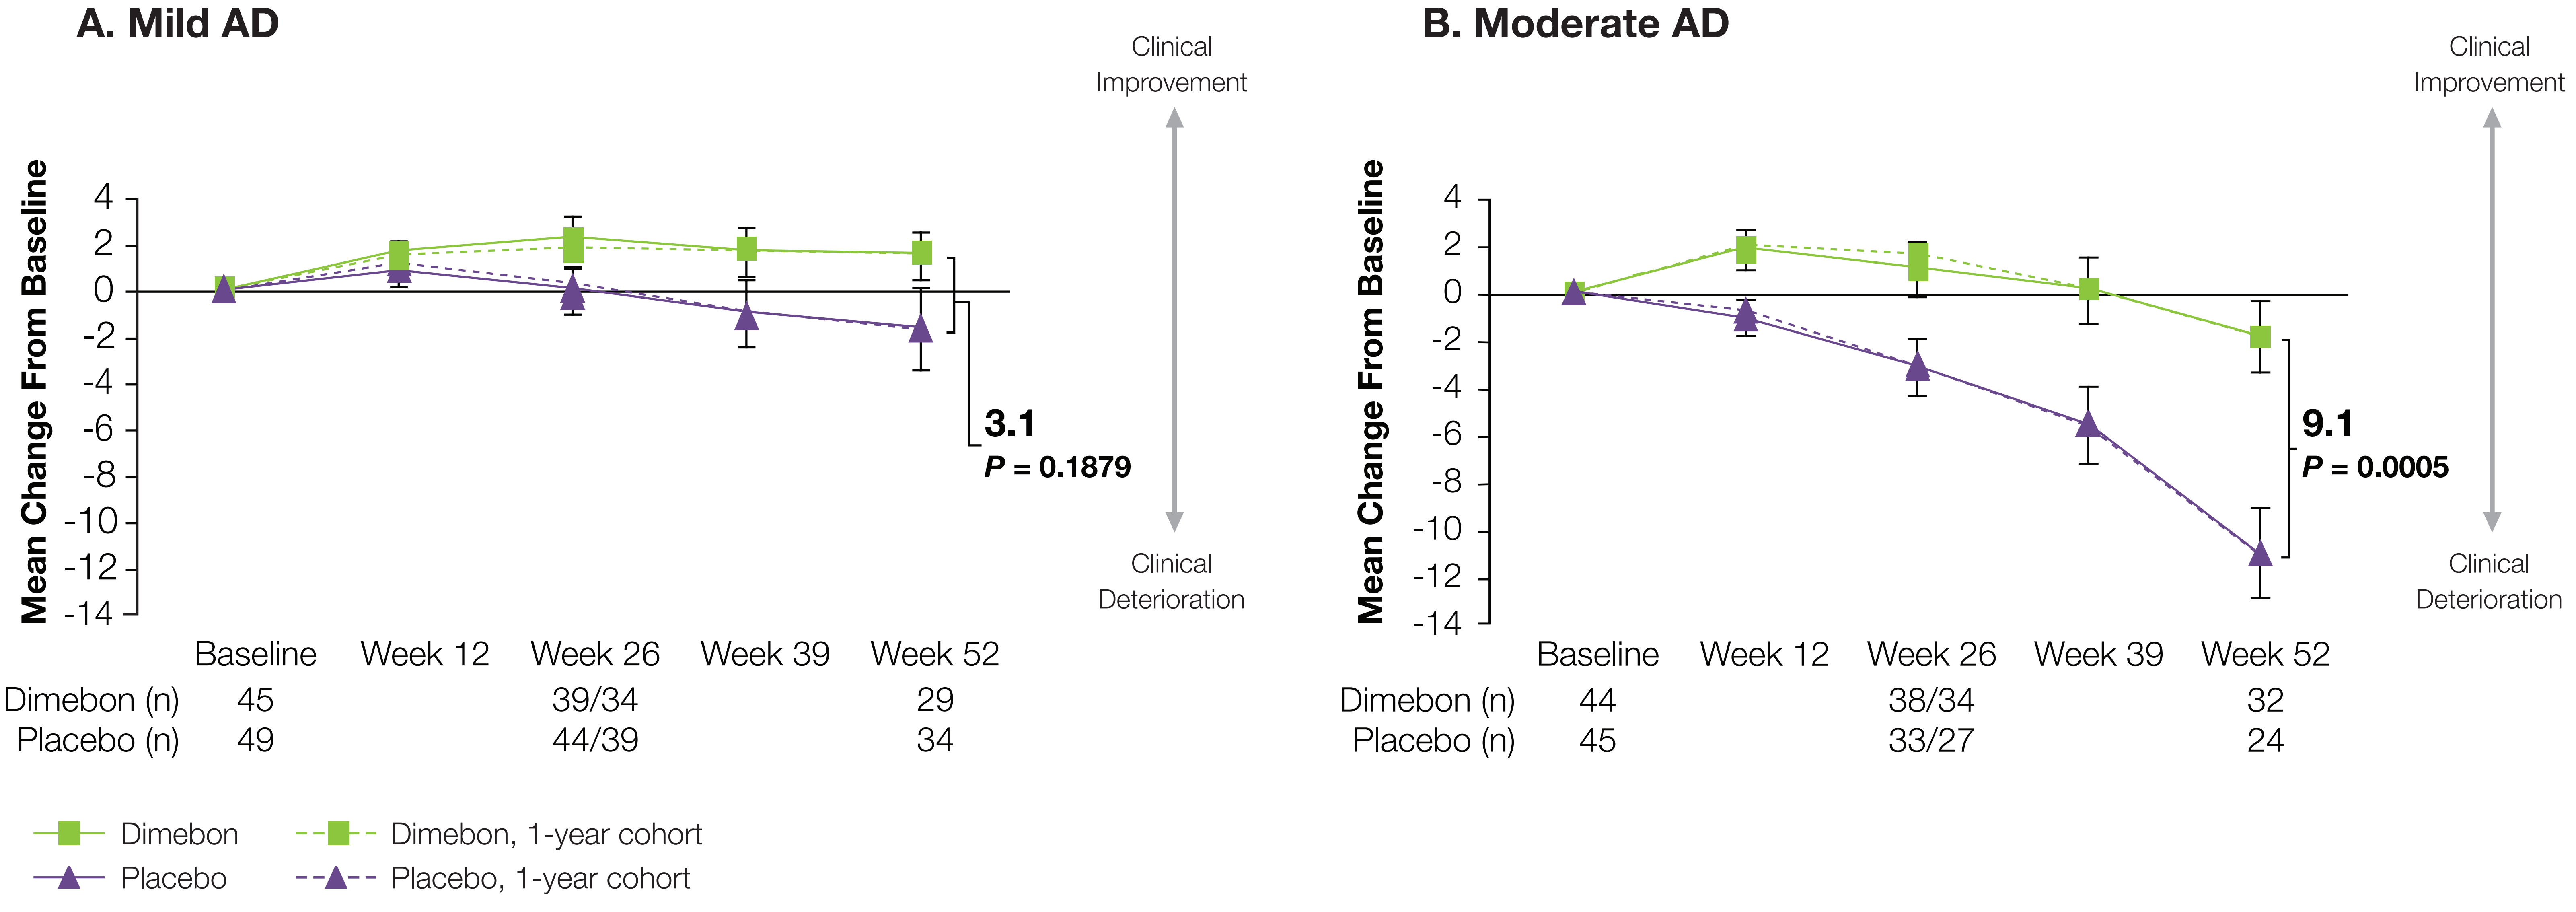
A. Mild AD
Mean Change From Baseline
4
2
0
-2
-4
-6
-8
-10
-12
-14
Baseline
Week 12
Week 26
Week 39
Week 52
Dimebon (n) 45 39/34 29
Placebo (n) 49 44/39 34
3.1
P = 0.1879
Clinical Improvement
Clinical Deterioration
B. Moderate AD
Mean Change From Baseline
4
2
0
-2
-4
-6
-8
-10
-12
-14
Baseline
Week 12
Week 26
Week 39
Week 52
Dimebon (n) 44 38/34 32
Placebo (n) 45 33/27 24
9.1
P = 0.0005
Clinical Improvement
Clinical Deterioration
Dimebon
Placebo
Dimebon, 1-year cohort
Placebo, 1-year cohort

Figure 1. CIBIC-plus: Dimebon Improved Overall Function at 1 Year in Patients With Mild and With Moderate AD

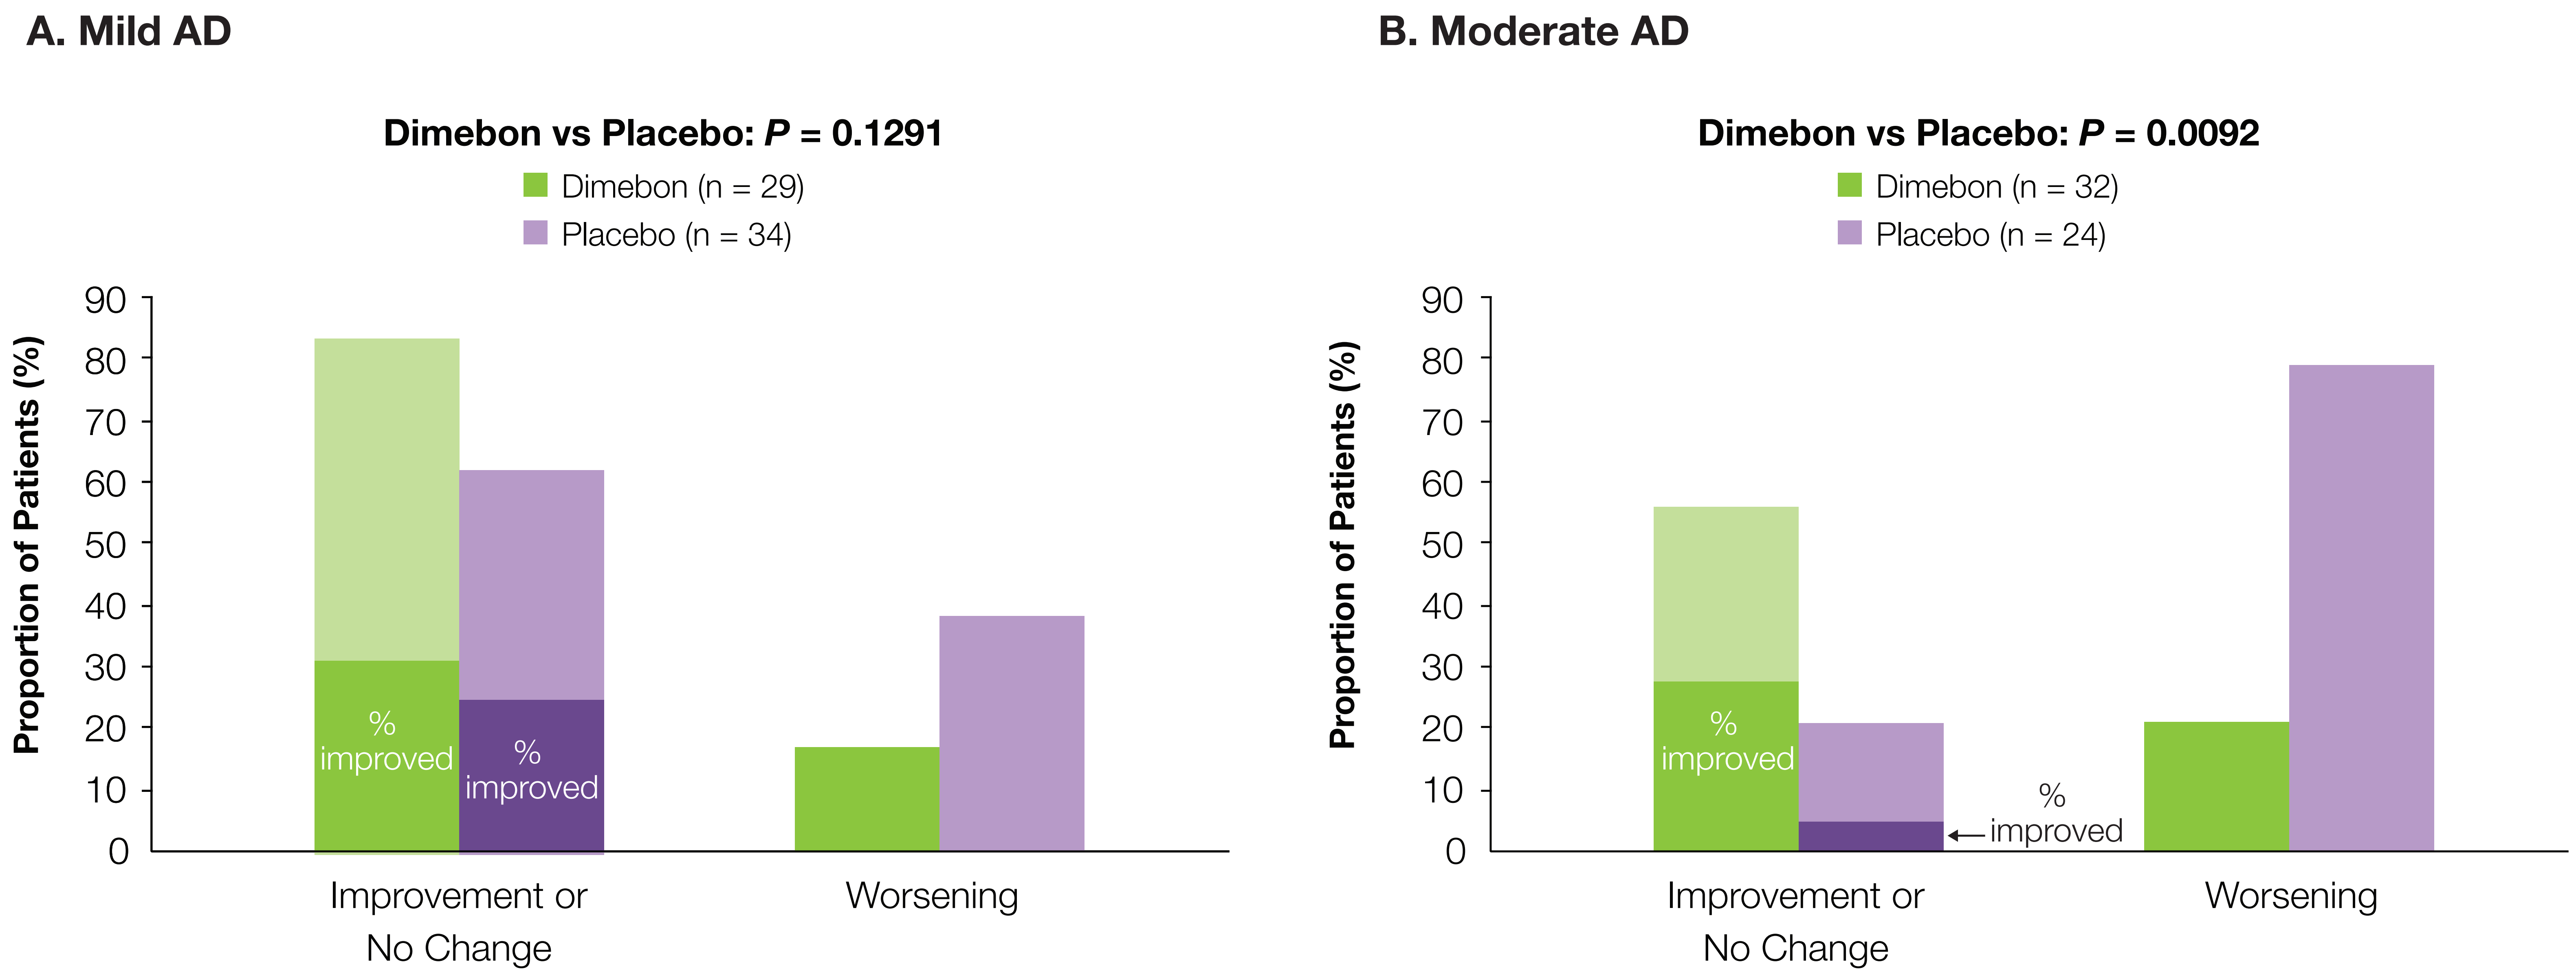

A. Mild AD
Dimebon vs Placebo: P = 0.1291
Dimebon (n = 29)
Placebo (n = 34)
Proportion of Patients (%)
90
80
70
60
50
40
30
20
10
0
%
improved
%
improved
Improvement or
No Change
Worsening
B. Moderate AD
Dimebon vs Placebo: P = 0.0092
Dimebon (n = 32)
Placebo (n = 24)
Proportion of Patients (%)
90
80
70
60
50
40
30
20
10
0
%
improved
%
improved
Improvement or
No Change
Worsening

Figure 3. NPI: Dimebon Significantly Improved Neuropsychiatric Symptoms in Patients With Moderate AD

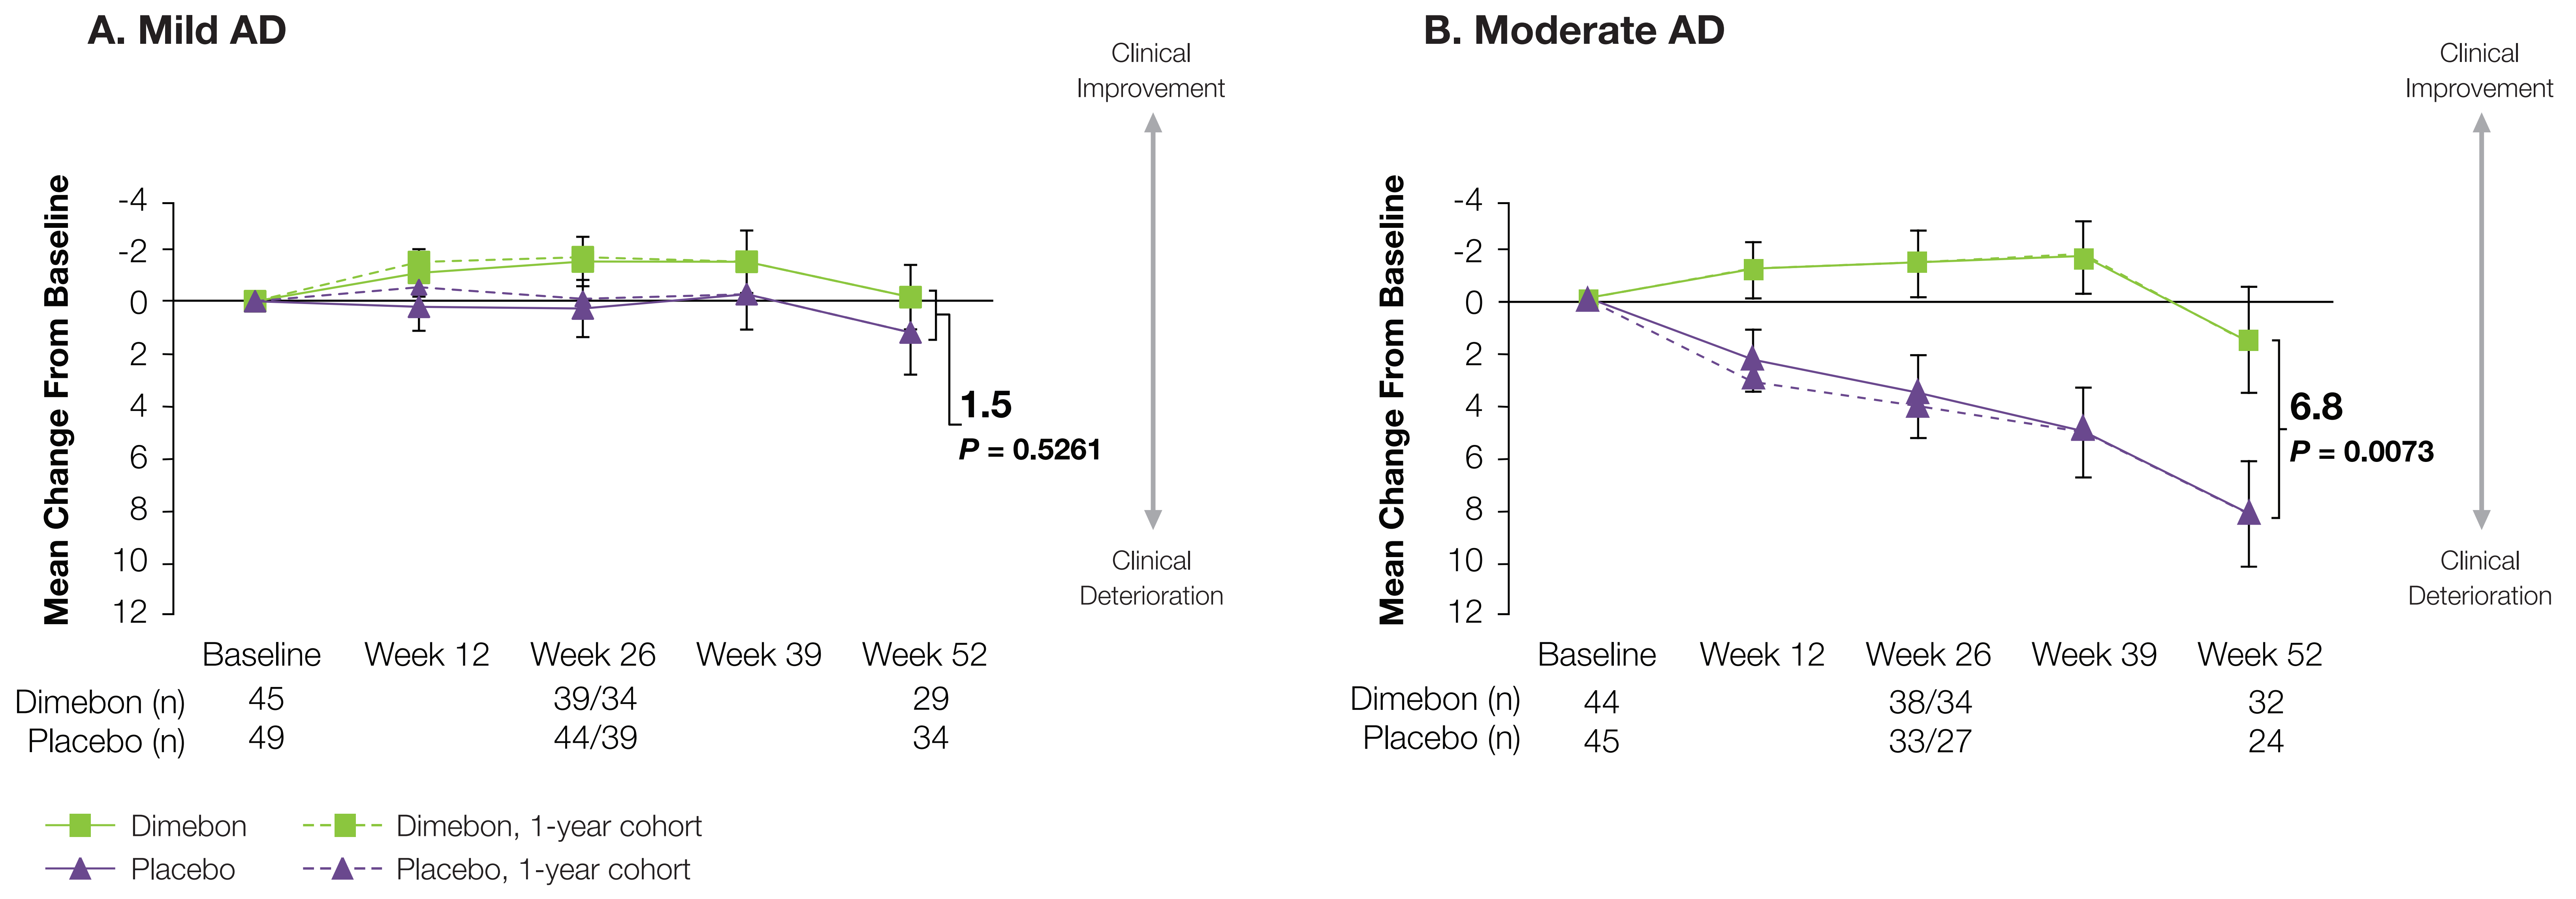

A. Mild AD
Clinical Improvement
Clinical Deterioration
Mean Change From Baseline
-4
-2
0
2
4
6
8
10
12
1.5
P = 0.5261
Baseline
Week 12
Week 26
Week 39
Week 52
Dimebon (n) 45 39/34 29
Placebo (n) 49 44/39 34
B. Moderate AD
Clinical Improvement
Clinical Deterioration
Mean Change From Baseline
6.8
P = 0.0073
Dimebon (n) 44 38/34 32
Placebo (n) 45 33/27 24
Dimebon
Dimebon, 1-year cohort
Placebo
Placebo, 1-year cohort

Table 1. Dimebon Treatment Was Well Tolerated

Week 52	Dimebon (n = 89)	Placebo (n = 94)
Number of patients reporting at least 1 adverse event, no. (%)	70 (79%)	70 (75%)
Number of subjects reporting at least 1 serious adverse event, no. (%)	3 (3%)	11 (12%)

CONCLUSIONS

- Patients with both mild and with moderate AD demonstrated benefit from treatment with Dimebon on cognition and memory, activities of daily living, behavior, and global function.
- Patients with mild AD were at or above baseline on all 5 outcome measures studied after 1 year of treatment.
 - Mild AD patients treated with Dimebon remained above baseline for 1 year on the ADAS-cog.
- Patients with moderate AD demonstrated substantial and sustained benefit over placebo on all 5 outcome measures at 1 year.
 - Moderate AD patients treated with Dimebon were improved 9.7 points over placebo on the ADAS-cog.
- Dimebon was safe and well tolerated.
- Dimebon improves the clinical course of patients with both mild and with moderate AD.

REFERENCE

1. Doody RS, Gavrilova SI, Sano M, et al; Dimebon investigators. Effect of dimebon on cognition, activities of daily living, behaviour, and global function in patients with mild-to-moderate Alzheimer's disease: a randomised, double-blind, placebo-controlled study. *Lancet*. 2008;372:207-215.