196

TETRABENAZINE FOR MODERATE VS. SEVERE/DISABLING CHOREA ASSOCIATED WITH HUNTINGTON'S DISEASE

Vivienne Shen, MD, PhD[1], Kathleen Clarence-Smith, MD, PhD[2], Christine Hunter, RN[3], Joseph Jankovic, MD[3]

[1]Lundbeck LLC, Deerfield, IL; [2]KM Pharmaceutical Consulting LLC, Washington, DC; [3]Baylor College of Medicine, Houston, TX

ABSTRACT (UPDATED)

Objective: To assess dosing, efficacy, and safety of tetrabenazine (TBZ) for patients with moderate vs. severe/disabling chorea associated with Huntington's disease (HD).

Background: At study initiation (1979), TBZ had not yet been approved in the US (2008).

Methods: In an open-label study, patients with hyperkinetic movement disorders were evaluated at the Parkinson's Disease Center and Movement Disorders Clinic (PDCMDC), Baylor College of Medicine. TBZ was used "last resort" when other medications failed to provide satisfactory symptom control. For HD-chorea patients, all previous chorea treatments were discontinued before TBZ initiation. Dosage was increased every 3 days, until a dosage-limiting AE occurred. TBZ was then down-titrated to optimal dosage. Visits were 6 weeks after hospitalization, and every 3 months thereafter. Responses were rated on a scale of 1–5 (1 = marked chorea reduction, excellent improvement in function; 2 = moderate chorea reduction, very good improvement in function; 3 = fair chorea improvement, only mild improvement in function; 4 = poor/no response for chorea/function; 5 = worsening chorea/some functional deterioration).[1]

Results: By 2004, 98 HD-chorea patients had participated. At baseline, 44 had moderate and 54 had severe/disabling chorea. 45% with moderate vs. 61% with severe/disabling chorea received TBZ >2 years. Average daily dosages (SD; range of mean doses) were 60.5 mg (25.9; 16.9–138.1) and 74.8 mg (45.0; 21.4–225.5) for moderate and severe/disabling chorea, respectively. On optimal dosages, 71% of moderate chorea patients achieved a "marked or moderate" response (any time point) vs. 78% for severe/disabling. The 5 most common AEs (moderate, severe/disabling) were somnolence (43%, 22%), insomnia (16%, 15%), depression (23%, 11%), akathisia (7%, 15%), and nervousness (7%, 13%).

Conclusions: TBZ dosing is highly individualized and independent of chorea severity. Responses to TBZ and AE rates were similar for patients grouped by baseline chorea severity.

BACKGROUND

- Prior to approval of tetrabenazine (TBZ) for the treatment of chorea associated with Huntington's disease (HD) in the United States, some US patients were able to obtain the drug from abroad, while others received TBZ under physician Investigational New Drug Applications (INDs)
- The Parkinson's Disease Center and Movement Disorder Clinic (PDCMDC) at Baylor College of Medicine administered TBZ under Dr. Jankovic's IND, issued in 1979[2,3]
- Patients were enrolled and treated at the PDCMDC under Protocol H-721, "Compassionate Use of TBZ in the Treatment of Hyperkinesias," a single-center, open-label, individualized-dosage study[2,3]
- Over 25 years (January 1979–February 2004), a total of 98 patients with HD-chorea were treated and had complete records available for analysis[4]
- At study entry, chorea severity ranged from moderate to severe/disabling
- Data from these patients was analyzed for various treatment effects

OBJECTIVE

- To assess dosing, efficacy, and safety of TBZ for patients with moderate vs. severe/disabling HD-chorea at baseline

METHODS

Patients

- Patients with hyperkinetic disorders underwent a detailed neurologic examination and a video recording designed to capture the phenomenology and severity of the disorder
- Patients were eligible for enrollment if their HD-chorea was functionally significant (i.e., chorea had to interfere with activities of daily living, occupational activities, and/or academic activities)
- Patients were also required to have failed available conventional treatments or report insufficient benefit from these treatments

Treatment

- TBZ treatment was initiated at a dosage of 12.5 mg/day; dosage was increased by 12.5-mg/day increments every 3–7 days until HD-chorea symptoms improved satisfactorily or a troublesome adverse event (AE) occurred. If a troublesome AE occurred, the dosage was down-titrated to the optimal dosage, defined as the dosage judged by the investigator to provide the greatest efficacy with minimal or tolerable AEs.[4]

Assessments

- Outpatient visits were scheduled 6 weeks after treatment initiation and every 3 months thereafter
- Dosage, efficacy, and AEs were collected at each clinic visit and entered on a Case Report Form (CRF)
- Response to treatment was rated on a scale of 1–5[1]
 - 1 = marked chorea reduction, excellent improvement in function
 - 2 = moderate chorea reduction, very good improvement in function
 - 3 = fair chorea improvement, only mild improvement in function
 - 4 = poor or no response for chorea and function
 - 5 = worsening chorea and some functional deterioration

Data Extraction

- Data were extracted from the Baylor CRFs into a database
- The data transfer was audited and the data analyzed

Statistical Analysis

- Descriptive statistics were employed to summarize demographic and illness characteristics, response to treatment, and AEs

RESULTS

Demographics and Patient Disposition

- Overall, no notable differences were observed between patients with moderate chorea at baseline and patients with severe/disabling chorea at baseline (Table 1)

Table 1. Demographics and Patient Disposition

Demographic	Moderate chorea at BL (N=44)	Severe/disabling chorea at BL (N=54)
Female, n (%)	21 (48)	37 (69)
Age, mean (range), years	53 (31–75)	56 (32–79)
Time since symptom onset, mean (range), years	7 (<1–20)	9 (<1–35)
Length of study participation, mean (range), years	3.0 (<1–11)	3.2 (<1–9)
Cumulative TBZ treatment duration >2 years, n (%)	20 (45)	33 (61)
Treatment status, n (%)		
Continuing treatment	9 (20)	10 (19)
Withdrawn from treatment	35 (80)	44 (81)
Death	0	6 (11)
Adverse event	9 (20)	8 (15)
Lack of efficacy	1 (2)	3 (6)
Disorder resolved spontaneously	0	1 (2)
Travel/financial reasons	1 (2)	6 (11)
Other	24 (55)	20 (37)

BL=baseline.

TBZ Dosage

- TBZ dosage was highly individualized and independent of chorea severity (Figure 1)
- The average daily dosage for patients with moderate chorea and severe/disabling chorea at baseline was 60.5 (±25.9) mg/day and 74.8 (±45.0) mg/day, respectively

Figure 1. Mean Dosage Distribution by Chorea Severity at Baseline

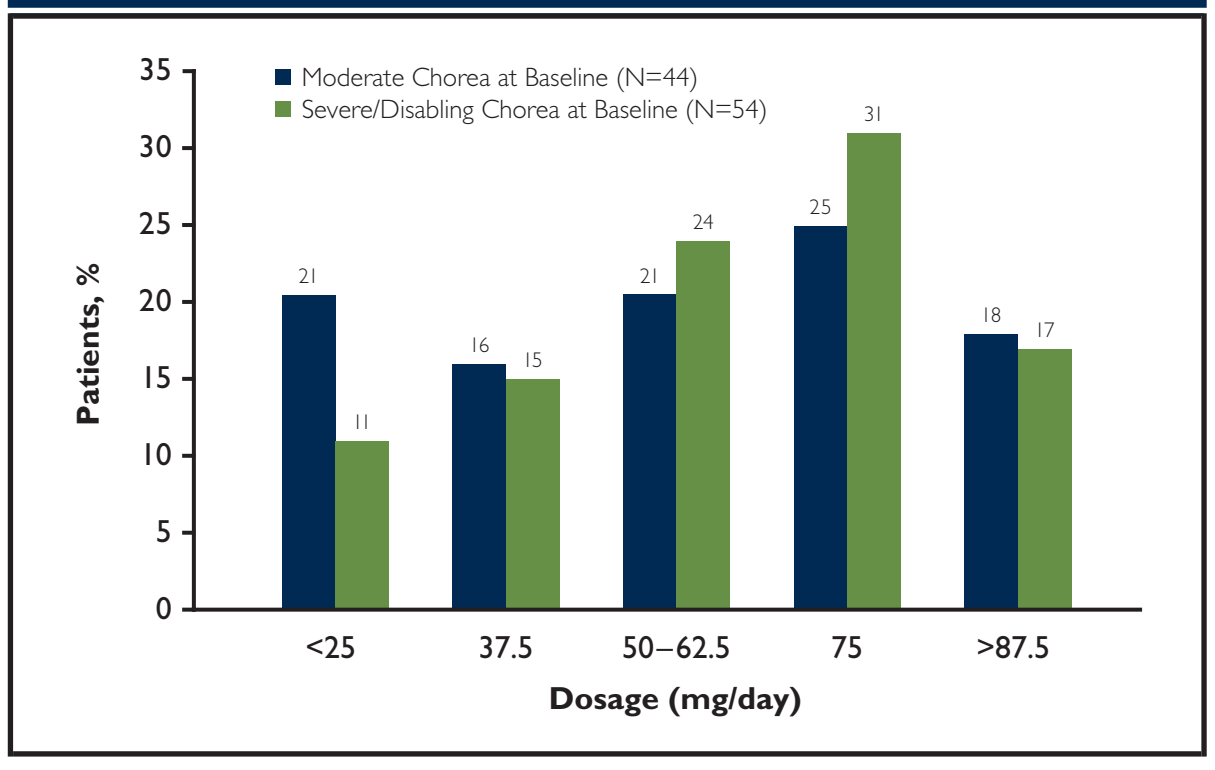
Moderate Chorea at Baseline (N=44)
Severe/Disabling Chorea at Baseline (N=54)
Patients, %
35
30
25
20
15
10
5
0
21
11
16
15
21
24
25
31
18
17
<25
37.5
50–62.5
75
>87.5
Dosage (mg/day)

Data is missing for 1 patient in the severe/disabling chorea at baseline group.

Efficacy

- At any dosage and time during the trial, 79% and 86% of patients with moderate chorea and severe/disabling chorea, respectively, achieved a "marked or moderate" response (Figure 2)

Figure 2. Efficacy Ratings of Patient Response to TBZ at Any Dosage at Any Time During the Trial

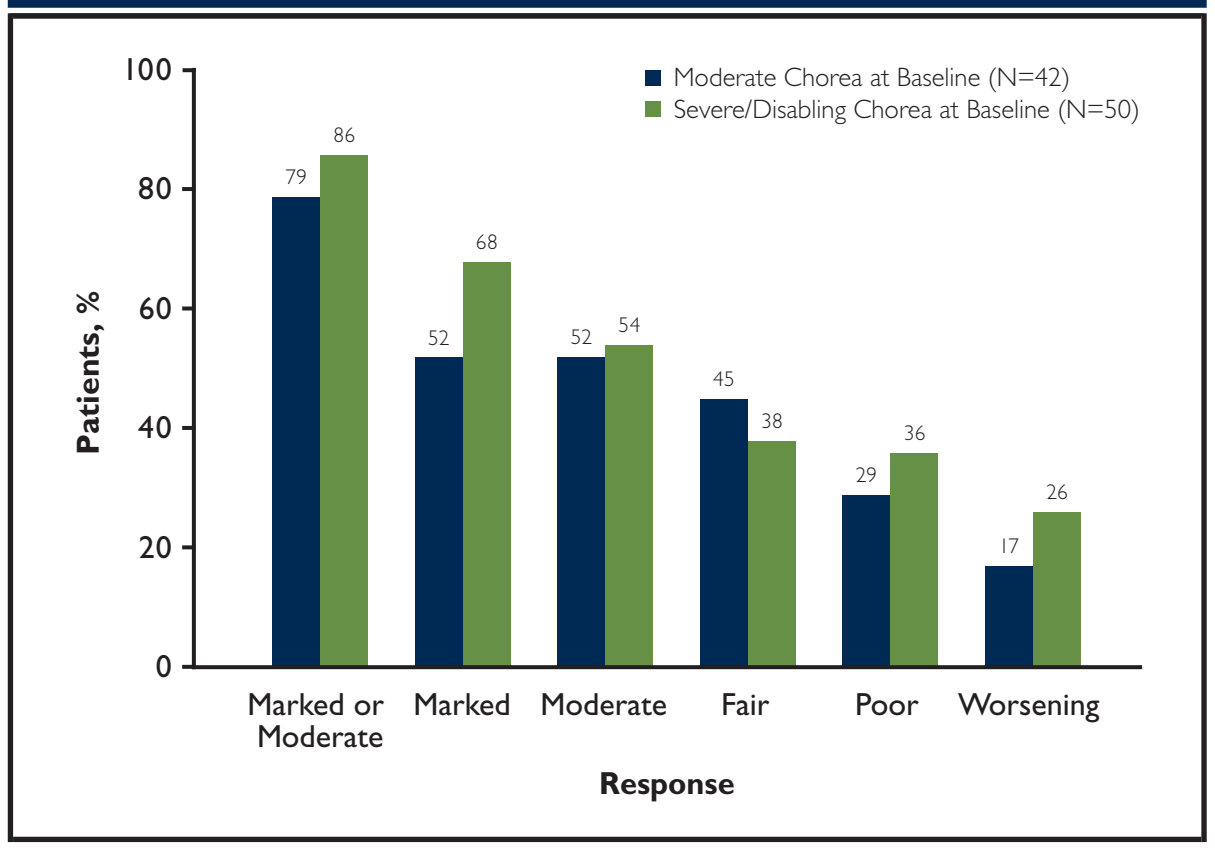
Moderate Chorea at Baseline (N=42)
Severe/Disabling Chorea at Baseline (N=50)
Patients, %
100
80
60
40
20
0
79
86
52
68
52
54
45
38
29
36
17
26
Marked or Moderate
Marked
Moderate
Fair
Poor
Worsening
Response

TBZ=tetrabenazine.
6 patients without valid response ratings are excluded.
Patients could be counted more than once.

- At optimal dosages at any time during the trial, 71% of patients with moderate chorea achieved a "marked or moderate" response compared with 78% for patients with severe/disabling chorea (Figure 3)

Figure 3. Patients With "Marked" and/or "Moderate" Response to TBZ at Optimal Dosage at Any Time During the Trial

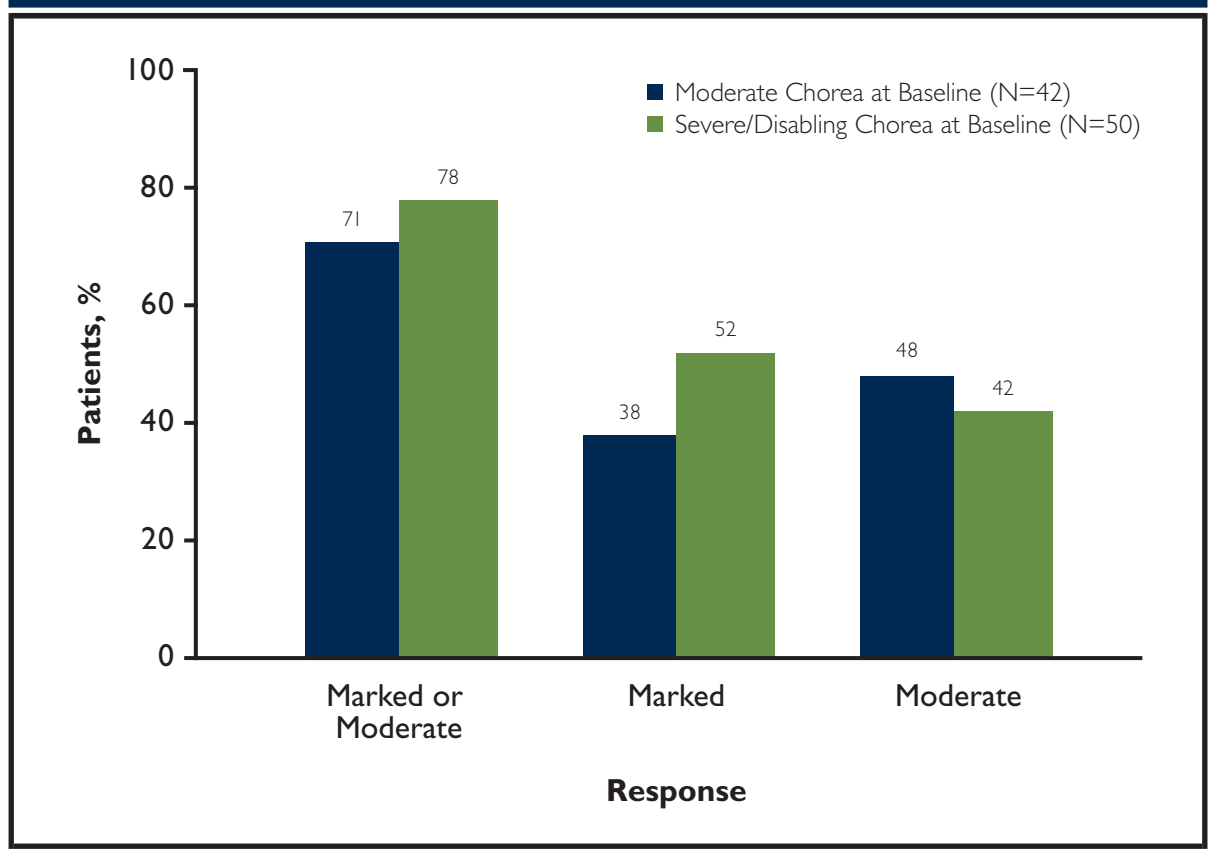
Moderate Chorea at Baseline (N=42)
Severe/Disabling Chorea at Baseline (N=50)
Patients, %
100
80
60
40
20
0
71
78
38
52
48
42
Marked or Moderate
Marked
Moderate
Response

TBZ=tetrabenazine.
6 patients without valid response ratings are excluded.
Patients could be counted more than once.

Safety

- The incidence of somnolence and depression for patients with moderate chorea at baseline (43% and 23%, respectively) was notably different from the incidence for patients with severe/disabling chorea at baseline (22% and 11%, respectively; Figure 4)

Figure 4. Incidence of Adverse Events[a] Following TBZ Treatment in Patients with Moderate or Severe/Disabling Chorea at Baseline

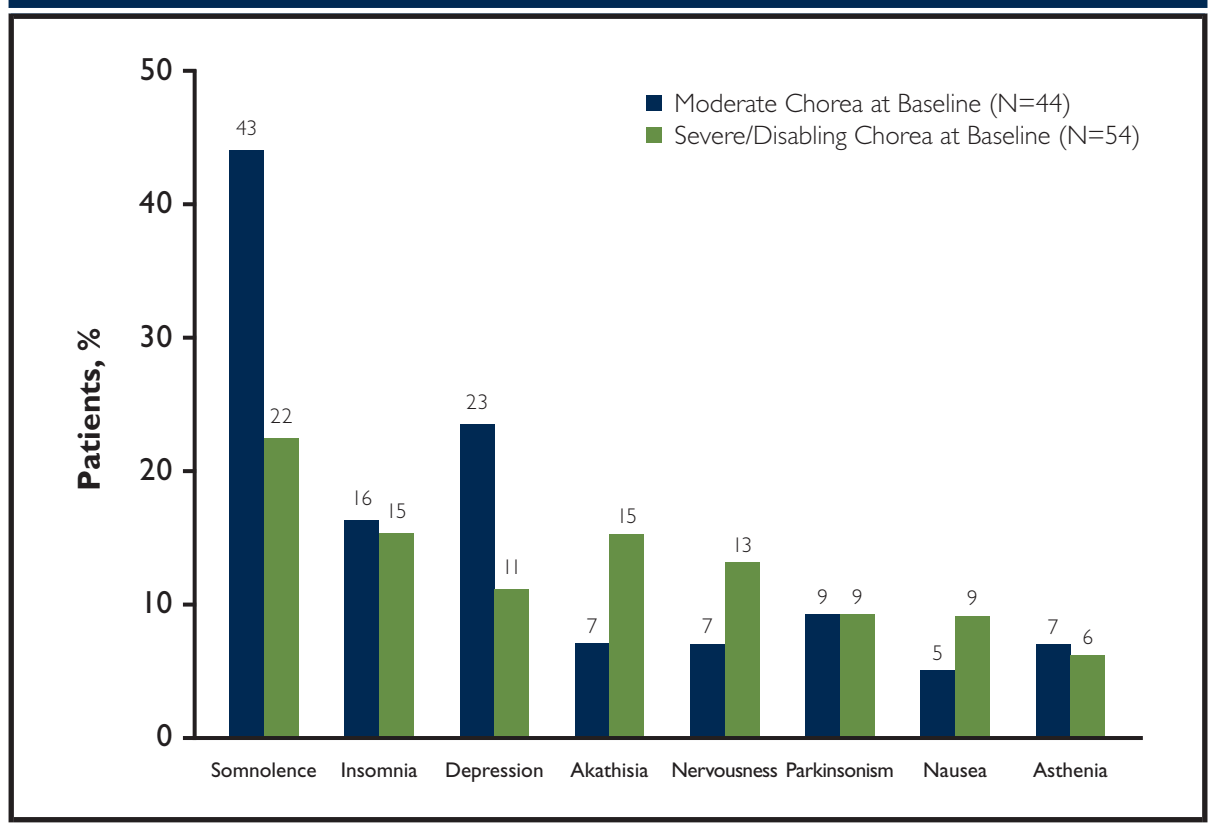
Moderate Chorea at Baseline (N=44)
Severe/Disabling Chorea at Baseline (N=54)
Patients, %
50
40
30
20
10
0
43
22
16
15
23
11
7
15
7
13
9
9
5
9
7
6
Somnolence
Insomnia
Depression
Akathisia
Nervousness
Parkinsonism
Nausea
Asthenia

TBZ=tetrabenazine.
[a]AEs were possibly or probably related to TBZ.

CONCLUSIONS

- TBZ dosing is highly individualized and independent of chorea severity
- Overall response to TBZ was similar between patients in both severity groups in this *post-hoc* analysis
- AE rates following TBZ treatment were similar between both baseline chorea severity patient groups

REFERENCES

1. Jankovic J, Orman J. *Neurology.* 1988;38:391–4.
2. Jankovic J, Beach J. *Neurology.* 1997;48:358–62.
3. Kenney C, Hunter C, Jankovic J. *Mov Disord.* 2007;22:193–7.
4. Jankovic J, Clarence-Smith K. *Expert Rev Neurother.* 2011;11:1509–23.

ACKNOWLEDGMENTS

The authors thank Statistics Collaborative, Inc. in Washington, DC, for providing the requested statistical summaries.

Presented at the Movement Disorder Society's (MDS) 16th International Congress of Parkinson's Disease and Movement Disorders, June 17–21, 2012, Dublin, Ireland
This study and analyses were funded by Lundbeck LLC, Deerfield, Illinois, USA. Medical writing and editorial assistance, provided by Prescott Medical Communications Group, Chicago, Illinois, was funded by Lundbeck LLC, Deerfield, Illinois, USA.