P04.230

Cervical Dystonia Patient Registry for Observation of BOTOX® Efficacy (CD PROBE) Interim Results of Pain-Related Outcomes

David Charles[1], Charles Adler[2], Mark Stacy[3], Cynthia Comella[4], Joseph Jankovic[5], Lee Ming Boo[6*], Mitchell Brin[6], Spyridon Papapetropoulos[6] (on behalf of the CD PROBE Study Group)

[1]Vanderbilt University Medical Center, Nashville, TN; [2]Mayo Clinic, Scottsdale, AZ; [3]Duke University Medical Center, Durham, NC; [4]Rush-Presbyterian-St Luke's Medical Center, Chicago, IL; [5]Baylor College of Medicine, Houston, TX; [6]Allergan, Inc., Irvine, CA

INTRODUCTION

- Cervical dystonia (CD) is one of the most common forms of adult-onset focal dystonia.
- CD is associated with abnormal contraction of neck muscles causing an abnormal head position and is commonly associated with pain.
- Pain associated with CD can lead to missed work and difficulties with activities of daily living.
- Neurotoxin injection is accepted as first line treatment for CD.[1]
- OnabotulinumtoxinA (BOTOX® Allergan Inc.) was the first neurotoxin approved by the US Food and Drug Administration for the treatment of CD.[2]
- Neurotoxin injection for CD has been utilized worldwide for more than 20 years.
- Physicians differ in the care of patients and in the application of neurotoxin injection for CD.
- CD-PROBE (Cervical Dystonia Patient Registry for Observation of BOTOX® Efficacy) is a multicenter, prospective, standard-of-care, observational registry designed to capture data on patients' clinical presentation, dosing of onabotulinumtoxinA, and treatment outcomes.

OBJECTIVE

- To report an interim analysis of pain-related outcomes in subjects receiving injections of onabotulinumtoxinA as part of the ongoing CD-PROBE clinical registry.

METHODS

- CD-PROBE is a prospective, open label, multi-center, clinical registry for subjects treated for CD (ClinicalTrials.gov Identifier NCT00836017).
- Subjects were administered 3 injections separated by >90 days. The dose and treatment intervals were based on standard of care for the physician's practice.
- Assessments made at baseline (injection 1) and 4-6 weeks after each injection (peak effect).
- Pain measures included the Pain Numeric Rating Scale (P-NRS), Toronto Western Spasmodic Torticollis Rating Scale (TWSTRS) pain subscale, and Cervical Dystonia Impact Profile (CDIP-58).
- Inclusion criteria:
 1. Subjects with CD appropriate for neurotoxin therapy
 2. Subjects are at least one of the following:
 a) New to the treating physician
 b) Not previously treated with neurotoxin
 c) If previously treated, have not received neurotoxin for ≥16 weeks
 3. Available to understand and participate in study procedures
 4. Provided informed consent and release of health information related to the study
- Exclusion criteria:
 1. A planned surgical procedure during the study
 2. Pregnancy or nursing, or a planned pregnancy during the study
 3. Poor compliance with past treatment plans
 4. Medical condition that could place the subject at risk if they participated in the study.

RESULTS

Table. Baseline Demographic and Disease Characteristics

Enrollment as of October 11, 2010, N	499
Sex, n (%)	
Females	384 (77.0)
Males	115 (23.0)
Race/ethnicity, n (%)	
Asian	10 (2.0)
Black	6 (1.2)
Hispanic	13 (2.6)
Native American	1 (0.2)
Other	1 (0.2)
White	468 (93.8)
Age, y, mean ± SD	57.5 ± 14.3
Height, in, mean ± SD	65.8 ± 7.7
Weight, lbs, mean ± SD	160.8 ± 38.6
BMI, kg/m², mean ± SD	26.3 ± 5.5
Age at symptom onset, y, mean ± SD	48.2 ± 16.1
Time to CD treatment after diagnosis, y, mean ± SD	1.0 ± 3.7
TWSTRS scores, mean ± SD	n=131
Severity	18.1 ± 5.0
Disability	11.5 ± 6.8
Pain	10.9 ± 5.3
Total	40.5 ± 13.2

CD = cervical dystonia; SD = standard deviation; TWSTRS = Toronto Western Spasmodic Torticollis Rating Scale.

OnabotulinumtoxinA dosage

- The mean ± SD dose at injection 1 (baseline) was 175.6 ± 104.9 U.
- The mean ± SD time interval between injections 1 and 2 was 100.4 ± 22.9 days and 100.0 ± 22.3 days between injections 2 and 3.
- At the time of this interim analysis, 495 patients received 1 injection, 312 received 2 injections, and 194 received 3 injections

Summary of pain improvement

- 89.6% (440/491) of subjects reported pain at baseline.
- Pain measured by the TWSTRS pain subscale significantly improved from 10.9 ± 5.3 at baseline to 7.5 ± 5.5 (*P*<0.0001) **(Figure 1).**
- Pain measured by P-NRS significantly improved from 6.1 ± 2.2 to 4.4 ± 2.7 (*P*<0.0001) **(Figure 2).**
- All domains for CDIP-58 improved significantly, including the pain subscale, which improved from 70.6 ± 24.7 to 52.0 ± 24.2 (*P*<0.0001) **(Figure 3).**

Figure 1. Toronto Western Spasmodic Torticollis Scores (TWSTRS), pain subscale (n=131)

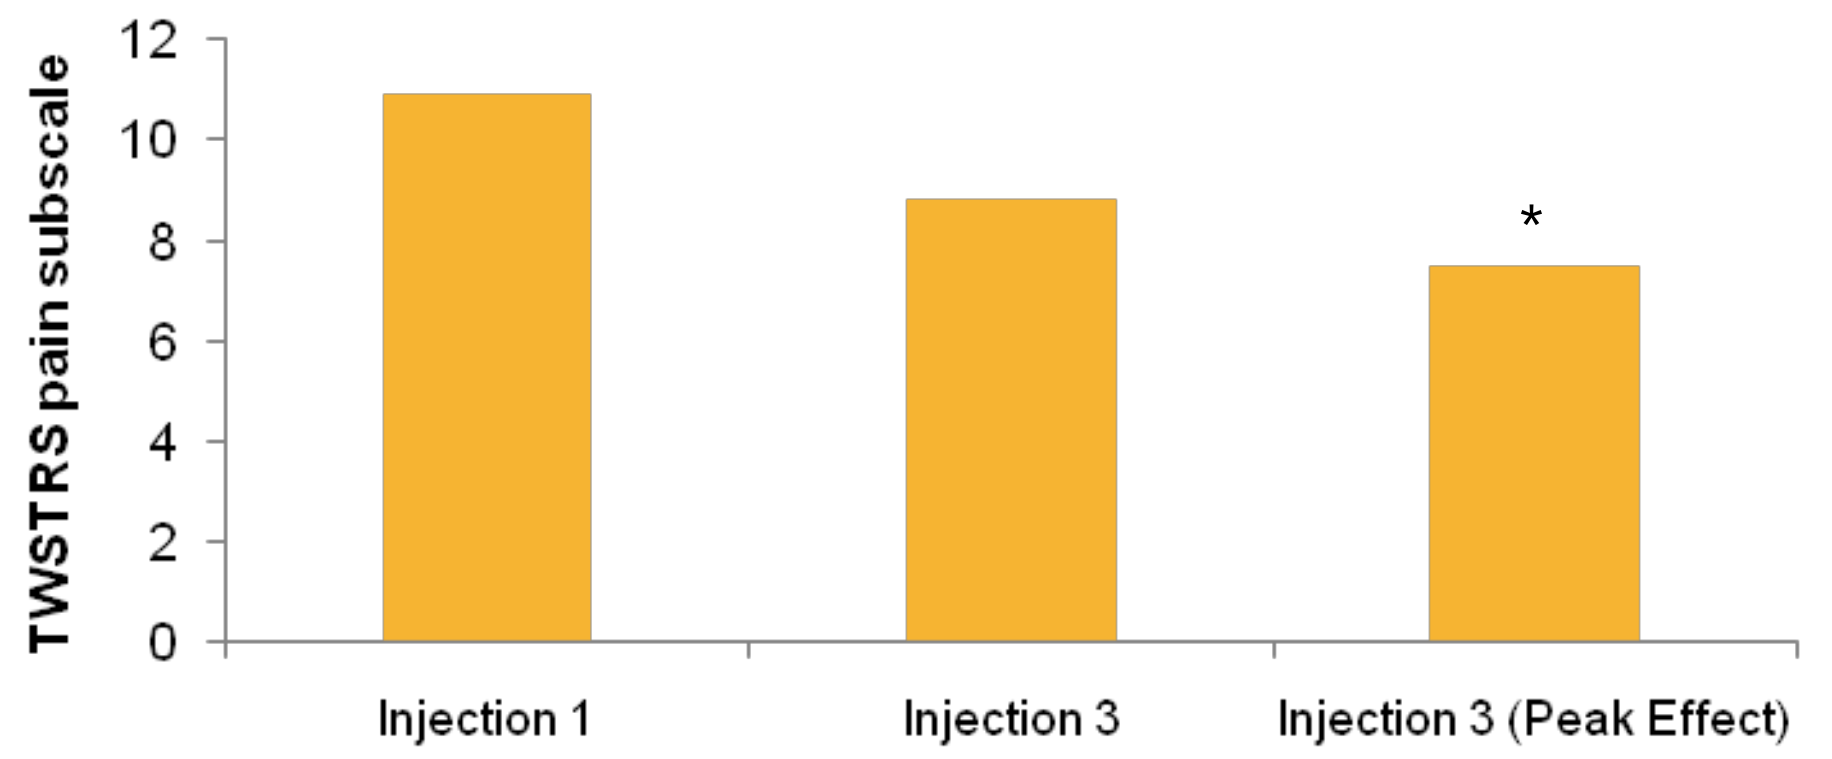
TWSTRS pain subscale
12
10
8
6
4
2
0
*
Injection 1
Injection 3
Injection 3 (Peak Effect)

*P<0.0001 vs baseline

Figure 2. Pain numeric rating scale (P-NRS) (n=84)

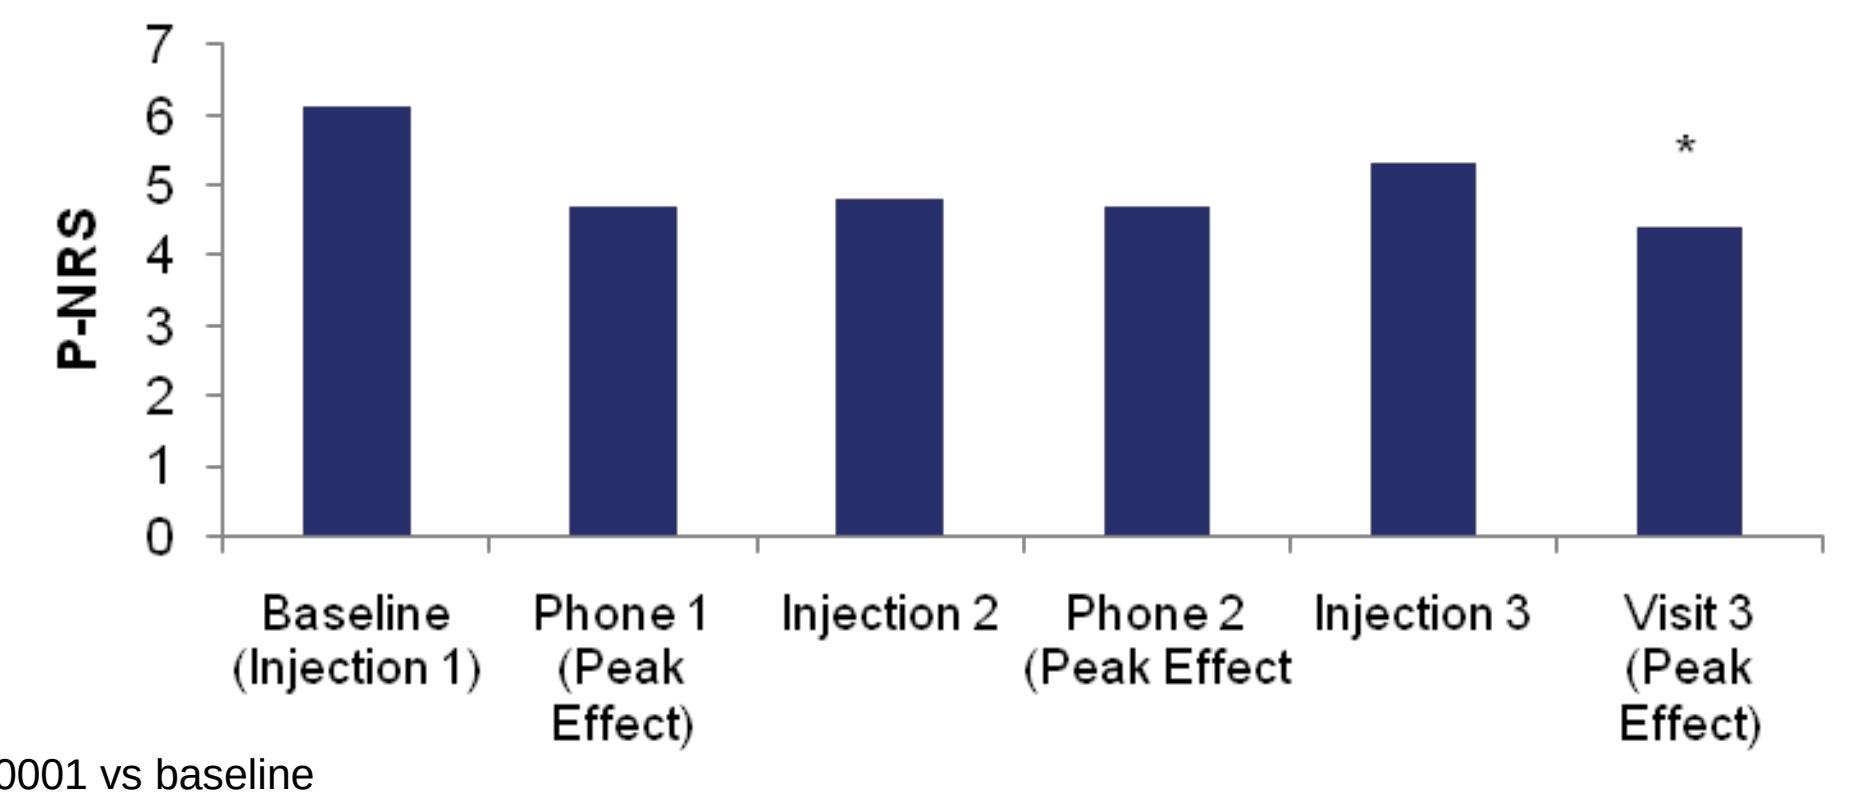
P-NRS
7
6
5
4
3
2
1
0
*
Baseline (Injection 1)
Phone 1 (Peak Effect)
Injection 2
Phone 2 (Peak Effect
Injection 3
Visit 3 (Peak Effect)

*P<0.0001 vs baseline

Figure 3. Cervical Dystonia Impact Profile (CDIP-58) (n=111)

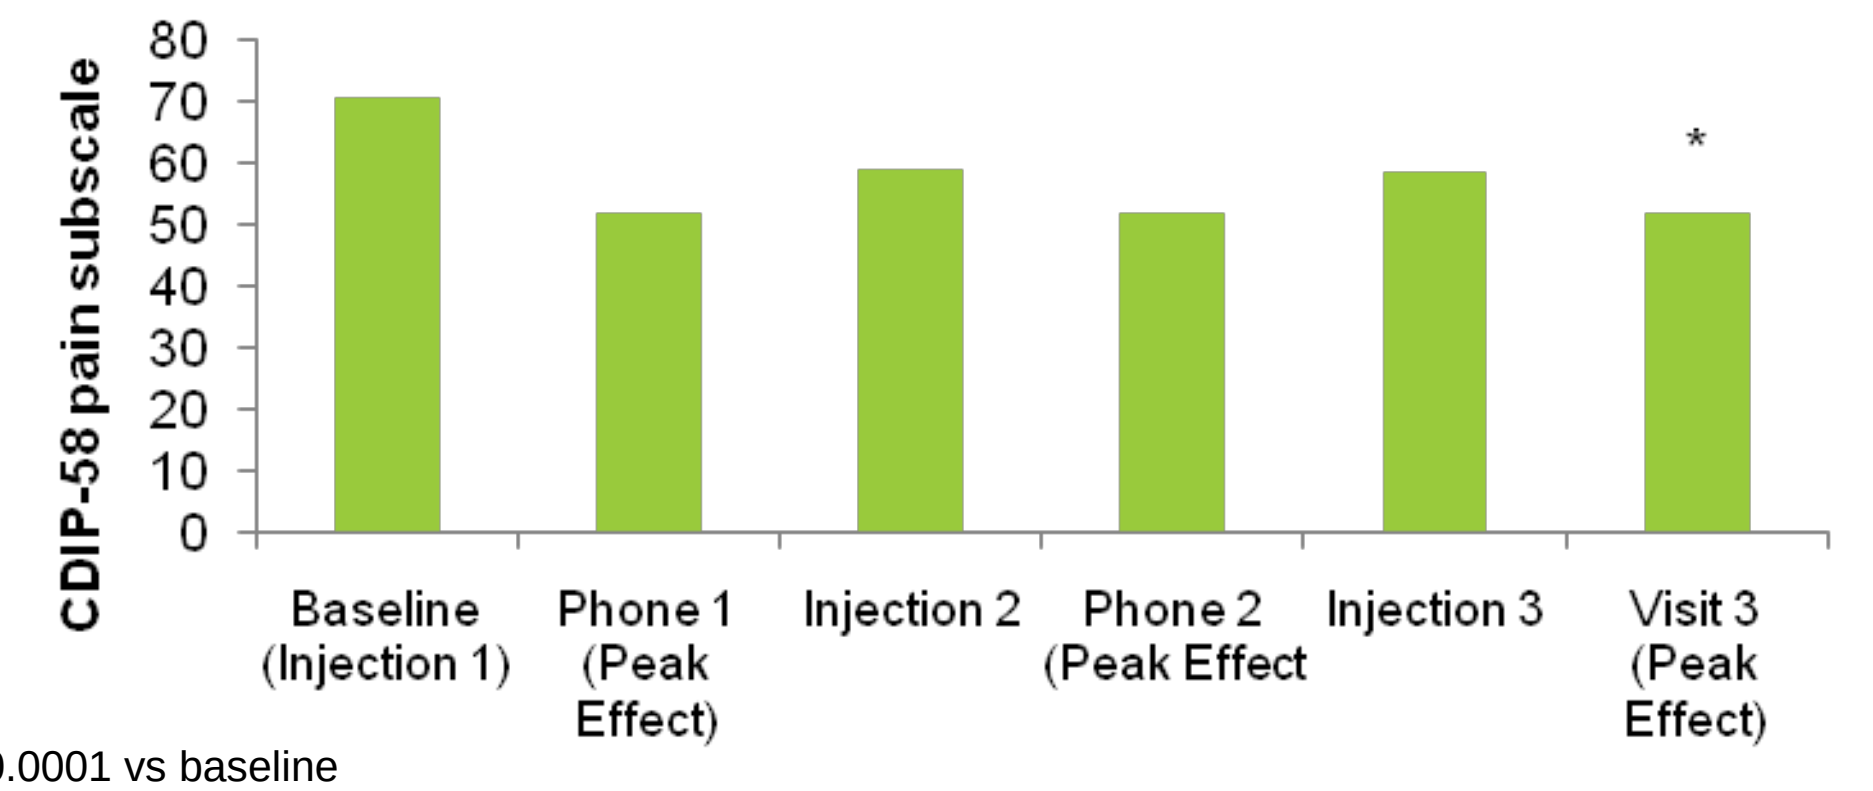
CDIP-58 pain subscale
80
70
60
50
40
30
20
10
0
*
Baseline (Injection 1)
Phone 1 (Peak Effect)
Injection 2
Phone 2 (Peak Effect
Injection 3
Visit 3 (Peak Effect)

*P<0.0001 vs baseline

Correlations of pain measures

- The TWSTRS pain subscale correlated with the CDIP-58 pain subscale at baseline (r=0.61; *P*≤0.0001).
- The TWSTRS pain subscale also correlated with the P-NRS at baseline (r=0.78; *P*≤0.0001).
- The CDIP-58 pain subscale correlated with the P-NRS at baseline (r=0.56; *P*≤0.0001).

CONCLUSIONS

- The majority of subjects with CD reported pain.
- The pain associated with CD improved significantly following onabotulinumtoxinA injection, as measured on 3 separate scales.
- There was a significant correlation of 3 measures used to assess pain at baseline: TWSTRS pain subscale, P-NRS, and CDIP-58 pain subscale.
- Additional information concerning the improvement of CD-associated pain relief following neurotoxin injection will become available as more subjects enroll in CD-PROBE.

References

1. Simpson DM, Blitzer A, Brashear A, et al. *Neurology*. 2008;70(19):1699-1706.
2. BOTOX® Prescribing Information. Irvine, CA: Allergan, Inc.; 2010.

CD PROBE Study Group

Lefko Aftonomos, CA; Pinky Agarwal, WA; Fahd Amjad, DC; Angela Applebee, VT; Kristin Appleby, DC; Richard Barbano, NY; Peter Barbour, PA; Brandon Barton, IL; Jay Bhatt, IN; Kevin Biglan, NY; David Bowers, TN; James Boyd, VT; Allison Brashear, NC; Mary Caire, TX; Barbara Changizi, NY; Mahan Chehrenama, VA; Shilpa Chitnis, TX; Cynthia Comella, IL; Boyd Crockett, MO; Paul Cullis, MI; Khashayar Dashtipour, CA; Lisa Davidson, MN; Thomas Davis, TN; J Antonelle De Marcaida, CT; Christina Drafta, NY; Drake Duane, AZ; Richard Dubinsky, KS; Jeffrey Esper, PA; Virgilio Evidente, AZ; Rez Farid, MO; Grace Forde, NY; Timothy Fries, VT; Ramon Gil, FL; John Goudreau, MI; David Greeley, WA; Alida Griffith, WA; Laurie Gutmann, WV; Gregory Hanes, FL; Robert Hauser, FL; Vanessa Hinson, SC; Patrick Hogan, WA; Tomas Holmlund, NY; Allan Ingenito, MN; Stuart Isaacson, FL; Bahman Jabbari, CT; Sandra Jacobson, AZ; Paul Jett, TN; John Kelemen, NY; Katie Kompoliti, IL; Daniel Kremens, PA; Rajeev Kumar, CO; Eugene Lai, TZ; Julie Leegwater-Kim, MA; Kenneth Levin, NJ; Peter LeWitt, MI; Tsao-Wei Liang, PA; Steven Lo, DC; Duarte Machado, CT; Padma Mahant, AZ; Zoltan Mari, MD; Anthony May, PA; Davis McDonald, FL; Stephen McGuire, TX; Emilio Melchionna, MA; Eric Molho, NY; Fatta Nahab, FL; Srinivas Nalamachu, KS; Anthony Nicholas, AL; Suneetha Nuthalapaty, TN; William Ondo, TX; Padraig O'Suilleabhain, TX; Fernando Pagan, DC; Atul Patel, KS; Gauri Pawar, WV; Diana Pollock, FL; Ben Renfroe, FL; Diana Richardson, CT; Perry Richardson, DC; Michael Rivner, GA; Jason Rosenberg, SC; David Ross, FL; Michael Rossen, MA; Kyle Ruffing, FL; Marwan Sabbagh, AZ; Johan Samanta, AZ; Cenk Sengun, FL; Aliya Sarwar, TX; Kapil Sethi, GA; Scott Sherman, AZ; Holly Shill, AZ; Tanya Simuni, IL; Carlos Singer, FL; Rodney Sorensen, WI; Michael Sorrell, MA; Natividad Stover, AL; Thyagarajan Subramanian, PA; William Sunter, FL; David Swope, CA; Tamara Miller, CO; Martin Taylor, OH; Margaret Tilton, NH; Bruno Tolge, NY; Richard Trosch, MI; Winona Tse, NY; Miodrag Velickovic, NY; Maureen Watts, TX; Robert Yapundich, NC; Cindy Zadikoff, IL; Lin Zhang, CA; Chong-Hao Zhao, CA

DISCLOSURE

This study and its analysis were sponsored by Allergan, Inc., Irvine, CA. Editing assistance was provided by Susan Sutch, PharmD, of Evidence Scientific Solutions, which was funded by Allergan, Inc.

Dr Charles receives income from Allergan Inc., Medtronic, and Pfizer for education and consulting services; Vanderbilt University received research support from Allergan Inc. and Medtronic for research lead by Dr Charles. Dr Adler received consulting fees from Eli Lilly, Ipsen, Medtronic, and Merck Serono. Dr Stacy received compensation from Allergan Inc., Boehringer-Ingelheim Inc., General Electric, Novartis, Osmotica, Synosia, Schering-Plough, GlaxoSmithKline, Teva, Biogen, Neurologix for consulting, speaker bureau, protocol steering committee, and/or safety monitoring board; he received royalties from Informa Press; and research support from Ceregene, IMPAX, Michael J. Fox Foundation, Neuraltus Novartis, Parkinson Study Group, and Schering-Plough. Dr Comella served as a consultant for Ipsen, Merz, Allergan Inc., UCB, and Esai; her institution received research support from Merz, Allergan, Ipsen, and Boehringer-Ingelheim Inc. in which she was a Principal Investigator. Dr Jankovic received compensation from Allergan Inc., Chelsea Therapeutics, EMD Serono, Merz Pharmaceuticals, Lundbeck Inc. and Teva for consulting services; he received compensation from Medlink: Neurology for serving as an editorial board member; he received research support from Allergan Inc., Allon Therapeutics, Ceregene Inc., Chelsea Therapeutics, Diana Helis Henry Medical Research Foundation, EMD Serono. Huntington's Disease Society of America, Huntington Study Group, Impax Pharmaceuticals, Ipsen Limited, Lundbeck Inc, Michael J Fox Foundation for Parkinson Research, Medtronic, Merz Pharmaceuticals, National Institutes of Health, National Parkinson Foundation, Neurogen, St. Jude Medical, Teva Pharmaceutical Industries Ltd, University of Rochester, and Parkinson Study Group. Dr Boo received personal compensation from Allergan as an employee; she is currently an employee of Johnson and Johnson. Dr Brin is an employee of Allergan, Inc. and receives salary, stock, and stock options from Allergan, Inc. Dr Papapetropoulos is an employee of Allergan, Inc. and receives salary, stock, and stock options from Allergan, Inc.; previously he was employed by Biogen Idec, Inc. and received stock options from Biogen Idec, Inc.

The potency units of onabotulinumtoxinA are specific to the preparation and assay method utilized. They are not interchangeable with other preparations of botulinum toxin products and, therefore, units of biological activity of onabotulinumtoxinA cannot be compared with or converted into units of any other botulinum toxin products assessed with any other specific assay method.

*Dr Boo was employed at Allergan Inc at the time that this study was initiated. She is currently employed at Biosense Webster, Diamond Bar, CA.

Presented at the 63rd Annual Meeting of the American Academy of Neurology (AAN), 9–16 April, 2011, Honolulu, Hawaii, USA.